Poster: 1655

ARV-393, a PROteolysis TArgeting Chimera (PROTAC) BCL6 Degrader, Combined With Biologics or Small-Molecule Inhibitors Induces Tumor Regressions in Diffuse Large B-Cell Lymphoma Models

Anna Van Acker, Lynn DeCarr, Sarah Eaton, Dan Sherman, Elizabeth Bortolon, Mark Bookbinder, Jennifer Pizzano, Morena Scopel, William Corwin, Ignacio Juncadella, Yongqing Huang, Ram Lingamaneni, Stephanie Renzullo, Jennifer Cantley, XiaoZhe (Janet) Wang, Ian Taylor, Sean Landrette, Sheryl M Gough

Arvinas Operations, Inc., New Haven, CT, USA

Objective

- To assess the activity of the PROTAC B-cell lymphoma 6 (BCL6) degrader, ARV-393, in combination with the standard of care (SOC) first-line chemotherapy regimen for diffuse large B-cell lymphoma (DLBCL), SOC biologics, or small-molecule inhibitors (SMIs) under clinical investigation in DLBCL xenograft models

Key Findings

- ARV-393 in combination with rituximab, cyclophosphamide, hydroxydaunorubicin, vincristine sulfate, and prednisone (R-CHOP), induced significantly greater tumor growth inhibition (TGI) compared with rituximab, CHOP, R-CHOP, or ARV-393 alone, with complete tumor regressions in all mice treated with the combination
- ARV-393 in combination with SOC biologics resulted in superior TGI compared with each agent alone, with complete tumor regressions observed in all mice treated with ARV-393 plus tafasitamab (anti–cluster of differentiation [CD]19) or rituximab (anti-CD20) and an increase in CD20 expression with ARV-393 alone
- ARV-393 in combination with investigational SMIs resulted in superior TGI compared with each agent alone, with tumor regressions observed in all mice treated with the combinations

Conclusions

- ARV-393 demonstrates synergistic antitumor activity, including complete regressions, in combination with SOC agents and select investigational SMIs in high-grade B-cell lymphoma (HGBCL) and aggressive DLBCL models
- These findings support future clinical investigation of ARV-393 in combination with SOC chemotherapy, SOC biologics, and investigational SMIs in patients with DLBCL
 - Preliminary studies demonstrating that ARV-393 increases CD20 expression provide additional support for the exploration of combinations with CD20-targeted agents and in the context of low or loss of CD20 expression

References

1. Leeman-Neill R and Bhagat G. Exp Opin Ther Targets. 2018;22(2):143-52.
2. Cattoretti G, et al. Cancer Cell. 2005;7(5):445-55.
3. Saito M, et al. Cancer Cell. 2007;12(3):280-92.
4. Sherman D, et al. Presented at AACR; April 5–10, 2024; San Diego, CA, USA. Abstract ND-05.
5. Gough S, et al. Poster presented at EHA; June 13–16, 2024; Madrid, Spain. Poster P1256 (QR code **Figure 2**).
6. Caimi PF, et al. Poster presented at AACR Advances in Malignant Lymphoma; June 19–22, 2024; Philadelphia, PA, USA. Poster PO-010.
7. Béguelin W, et al. Cancer Cell. 2016;30(2):197-213.

Disclosure

All authors are employees and shareholders of Arvinas Operations, Inc. Dan Sherman and Sheryl M Gough additionally hold patents with Arvinas Operations, Inc.

Acknowledgments

This research was funded by Arvinas, Inc. Medical writing support was provided by Lauren Hanlon, PhD, CMPP, of Red Nucleus and was funded by Arvinas Operations, Inc.

Contact

Anna Van Acker; anna.chiarella@arvinas.com

American Association for Cancer Research (AACR) Annual Meeting
Chicago, IL, USA; April 25–30, 2025

Background

- BCL6 is a preclinically validated oncogenic driver of DLBCL historically considered to be undruggable[1-3]
- Given the heterogeneity and multiple resistance mechanisms of DLBCL and that BCL6 regulates hundreds of genes linked to oncogenesis and resistance,[1] BCL6 degradation has the potential for broad drug combinability
- ARV-393, a PROTAC BCL6 degrader, directly binds an E3 ubiquitin ligase and BCL6 to induce the ubiquitination of BCL6 and its subsequent proteasomal degradation (**Figure 1**)[4]
- ARV-393 rapidly degrades BCL6 in DLBCL cell lines (>90% degradation in 2 hours), and its iterative activity overcomes rapid BCL6 resynthesis (**Figure 2**); single-agent ARV-393 induced potent TGI, including regressions, in DLBCL patient-derived xenograft models[5]
- ARV-393 monotherapy is being evaluated in a phase 1 trial (NCT06393738) in patients with non-Hodgkin lymphoma, including DLBCL[6]
- Here, we explore the preclinical efficacy of ARV-393 in combination with SOC therapies and SMIs targeting complementary mechanistic pathways in DLBCL

Figure 1: Mechanism of action of ARV-393

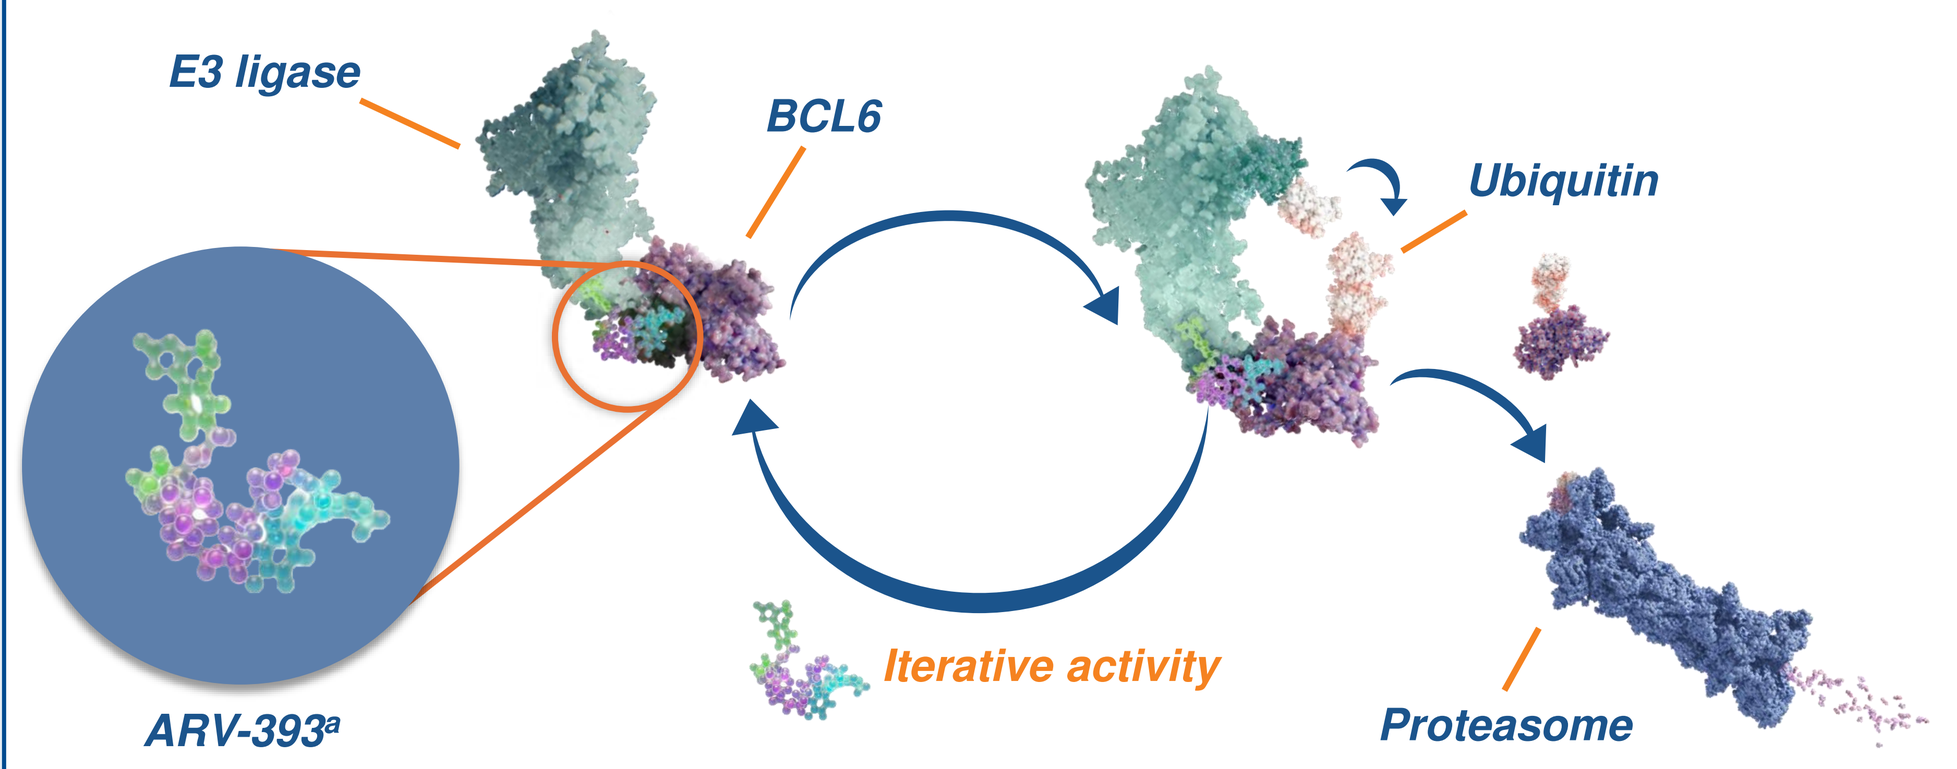
E3 ligase
BCL6
Ubiquitin
Iterative activity
Proteasome
ARV-393[a]

[a]General PROTAC protein degrader is shown.
BCL6=B-cell lymphoma 6; PROTAC=PROteolysis TArgeting Chimera.

Figure 2: ARV-393 potently and rapidly degrades BCL6 in vitro, critical to overcoming BCL6 resynthesis

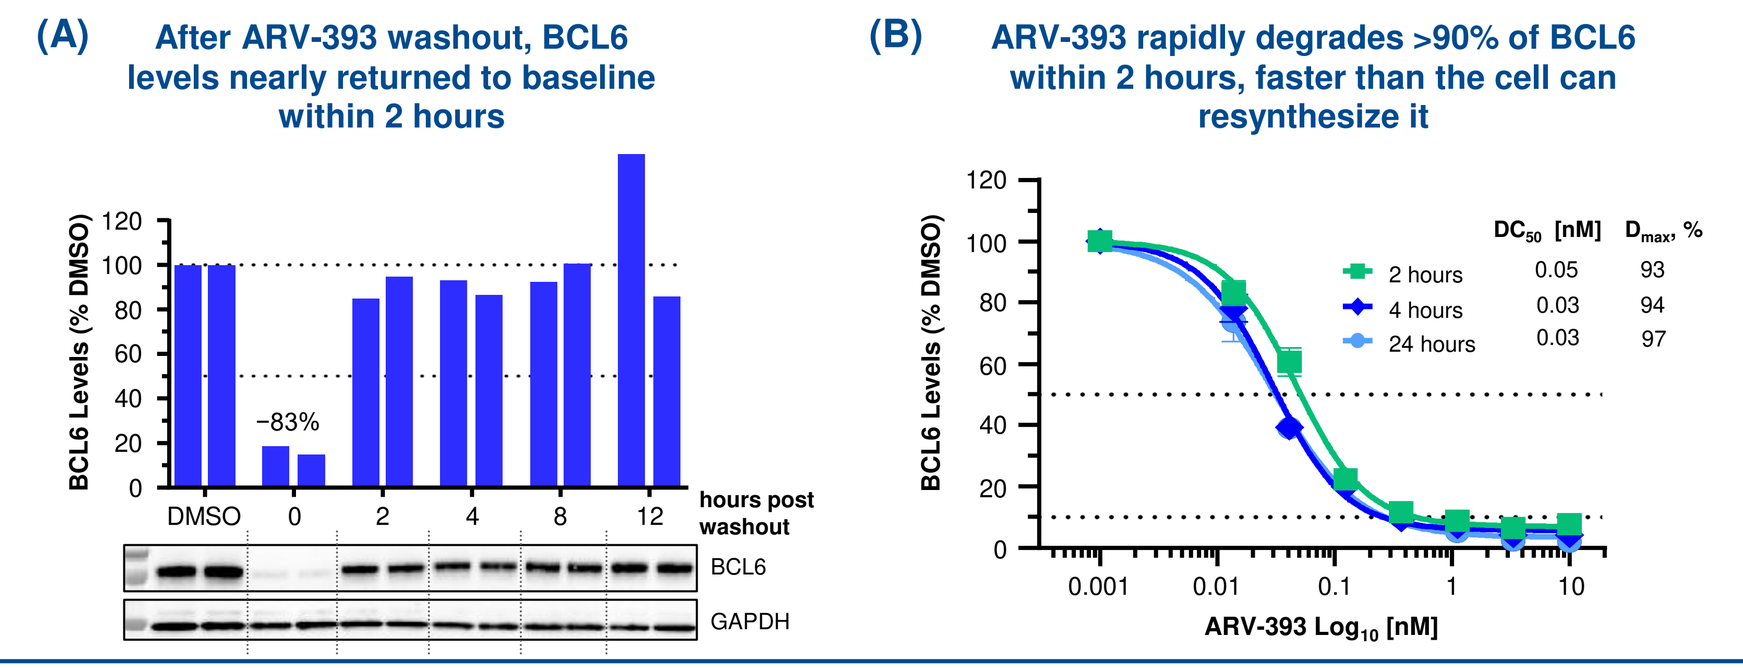
(A) After ARV-393 washout, BCL6 levels nearly returned to baseline within 2 hours
BCL6 Levels (% DMSO)
−83%
DMSO 0 2 4 8 12 hours post washout
BCL6
GAPDH
(B) ARV-393 rapidly degrades >90% of BCL6 within 2 hours, faster than the cell can resynthesize it
BCL6 Levels (% DMSO)
DC50 [nM] Dmax, %
2 hours 0.05 93
4 hours 0.03 94
24 hours 0.03 97
ARV-393 Log10 [nM]

Please scan this QR code to view **previously presented ARV-393 preclinical data.**[5]

(A) BCL6 protein levels quantified by Western blot in the DLBCL OCI-Ly1 cell line, following 4-hour treatment with ARV-393 1.5 nM. Duplicate samples shown following ARV-393 washout and addition of cereblon ligand to block residual ARV-393. **(B)** BCL6 degradation time-course in OCI-Ly1 cells.
BCL6=B-cell lymphoma 6; DC_{50}=half-maximal degradation concentration; DLBCL=diffuse large B-cell lymphoma; D_{max}=maximum level of degradation; DMSO=dimethyl sulfoxide; GAPDH=glyceraldehyde-3-phosphate dehydrogenase.

Methods

ARV-393 in Combination With R-CHOP

- A SU-DHL-4 cell line–derived xenograft (CDX) mouse model representing a HGBCL (with *MYC*, B-cell lymphoma 2 [*BCL2*], and *BCL6* rearrangements) was used to evaluate ARV-393 in combination with rituximab, CHOP, and R-CHOP
 - ARV-393 6 mg/kg or 30 mg/kg was administered orally (PO) once daily (QD) for 28 days; rituximab 3 mg/kg was administered intravenously (IV) on days 1, 8, 15, and 22; CHOP (30:2.475:0.375:0.15 mg/kg) was given IV on day 1 (prednisone was given PO QD on days 1–5); and R-CHOP followed these same dosing methods. The ARV-393 6 mg/kg dose was used for combination studies
 - Control groups included mice that received an immunoglobulin G1 (IgG1) IV on days 1, 8, 15, and 22 or mice treated with the oral vehicle QD

ARV-393 in Combination With SOC Biologics

- Using the SU-DHL-4 CDX mouse model, ARV-393 was evaluated in combination with clinically relevant doses of SOC biologic therapies
 - ARV-393 6 mg/kg PO QD was administered alone or in combination with tafasitamab (anti-CD19 biologic), polatuzumab vedotin (anti-CD79b antibody-drug conjugate), or rituximab
 - Tafasitamab 10 mg/kg was administered IV on days 1, 4, 8, 15, and 22; polatuzumab vedotin 1 mg/kg was administered IV on day 1; and rituximab 3 mg/kg or 10 mg/kg was administered IV on days 1, 8, 15, and 22
 - Control groups included mice that received IgG1 IV on days 1, 8, 15, and 22; mice treated with the oral vehicle QD; and mice that received lenalidomide 10 mg/kg PO QD combined with tafasitamab

ARV-393 in Combination With SMIs

- ARV-393 was evaluated in combination with SMIs in HGBCL or aggressive activated B-cell (ABC) DLBCL CDX models
 - ARV-393 30 mg/kg PO QD was administered alone or in combination with acalabrutinib (Bruton tyrosine kinase [BTK] SMI) or tazemetostat (EZH2 SMI); ARV-393 3 mg/kg PO QD was administered alone or in combination with venetoclax (BCL2 SMI)
 - Acalabrutinib 2 mg/kg PO was administered twice daily (BID) to mice bearing the ABC OCI-Ly10 *MYD88*-positive CDX, venetoclax 100 mg/kg PO QD to mice bearing the *BCL2*-positive OCI-Ly1 CDX, and tazemetostat 300 mg/kg PO BID to mice bearing the *EZH2*-mutant SU-DHL-6 HGBCL CDX
 - One group of mice from each model received the oral vehicle QD

Results

ARV-393 in Combination With R-CHOP

- The combination of ARV-393 with rituximab, CHOP, or R-CHOP (the first-line SOC therapy for DLBCL) all resulted in tumor regressions; ARV-393 combined with R-CHOP induced complete regressions and had significantly higher TGI compared with rituximab, CHOP, R-CHOP, or ARV-393 alone (**Figure 3**)
 - ARV-393 induced complete regressions in 4/10 mice when combined with rituximab, in 6/10 mice when combined with CHOP, and in 10/10 mice when combined with R-CHOP
 - Body weights were maintained with monotherapy and combination treatments

ARV-393 in Combination With SOC Biologics

- The combination of ARV-393 with SOC biologics targeting CD19 (tafasitamab), CD79b (polatuzumab vedotin), or CD20 (rituximab) resulted in tumor regressions and demonstrated significantly stronger TGI compared with either agent alone (**Figure 4**)
 - ARV-393 combined with tafasitamab induced complete regressions in 10/10 mice (**Figure 4A**)
 - In contrast, tafasitamab combined with lenalidomide resulted in 55% TGI
 - ARV-393 combined with polatuzumab vedotin induced complete regressions in 4/10 mice (**Figure 4B**)
 - ARV-393 combined with rituximab 3 mg/kg or 10 mg/kg induced complete regressions in 9/10 and 9/9 mice, respectively; of note, ARV-393 monotherapy resulted in a significant increase in CD20 expression compared with vehicle (**Figure 4C**)
 - Body weights were maintained with monotherapy and combination treatments

ARV-393 in Combination With SMIs

- The combination of ARV-393 with SMIs of BTK (acalabrutinib), BCL2 (venetoclax), or EZH2 (tazemetostat) demonstrated strong TGI, including tumor regressions in all mice (**Figure 5**)
 - ARV-393 combined with acalabrutinib showed significantly stronger TGI than either agent alone (**Figure 5A**)
 - ARV-393 combined with venetoclax demonstrated significantly stronger TGI compared with ARV-393 alone, whereas venetoclax monotherapy resulted in rebound of tumor growth and progressive disease (**Figure 5B**)
 - ARV-393 combined with tazemetostat showed significantly stronger TGI than either ARV-393 or tazemetostat monotherapy (**Figure 5C**), consistent with literature reports showing that BCL6 and EZH2 play cooperative roles in lymphomagenesis[7]
 - In this model, MYC, EZH2, and BCL2 protein levels were increased by 56%, 66%, and 12%, respectively, with ARV-393 alone vs vehicle, but were decreased by 75%, 80%, and 96%, respectively, with ARV-393 plus tazemetostat vs vehicle, demonstrating a synergistic reduction in proteins known to drive lymphoma cell growth
 - BCL6 degradation was greater with ARV-393 combined with tazemetostat vs ARV-393 alone (87% vs 65%)
 - Body weights were maintained with monotherapy and combination treatments, with dosing holidays implemented in the venetoclax and tazemetostat combinations

Figure 3: ARV-393 in combination with R-CHOP (SU-DHL-4 model)

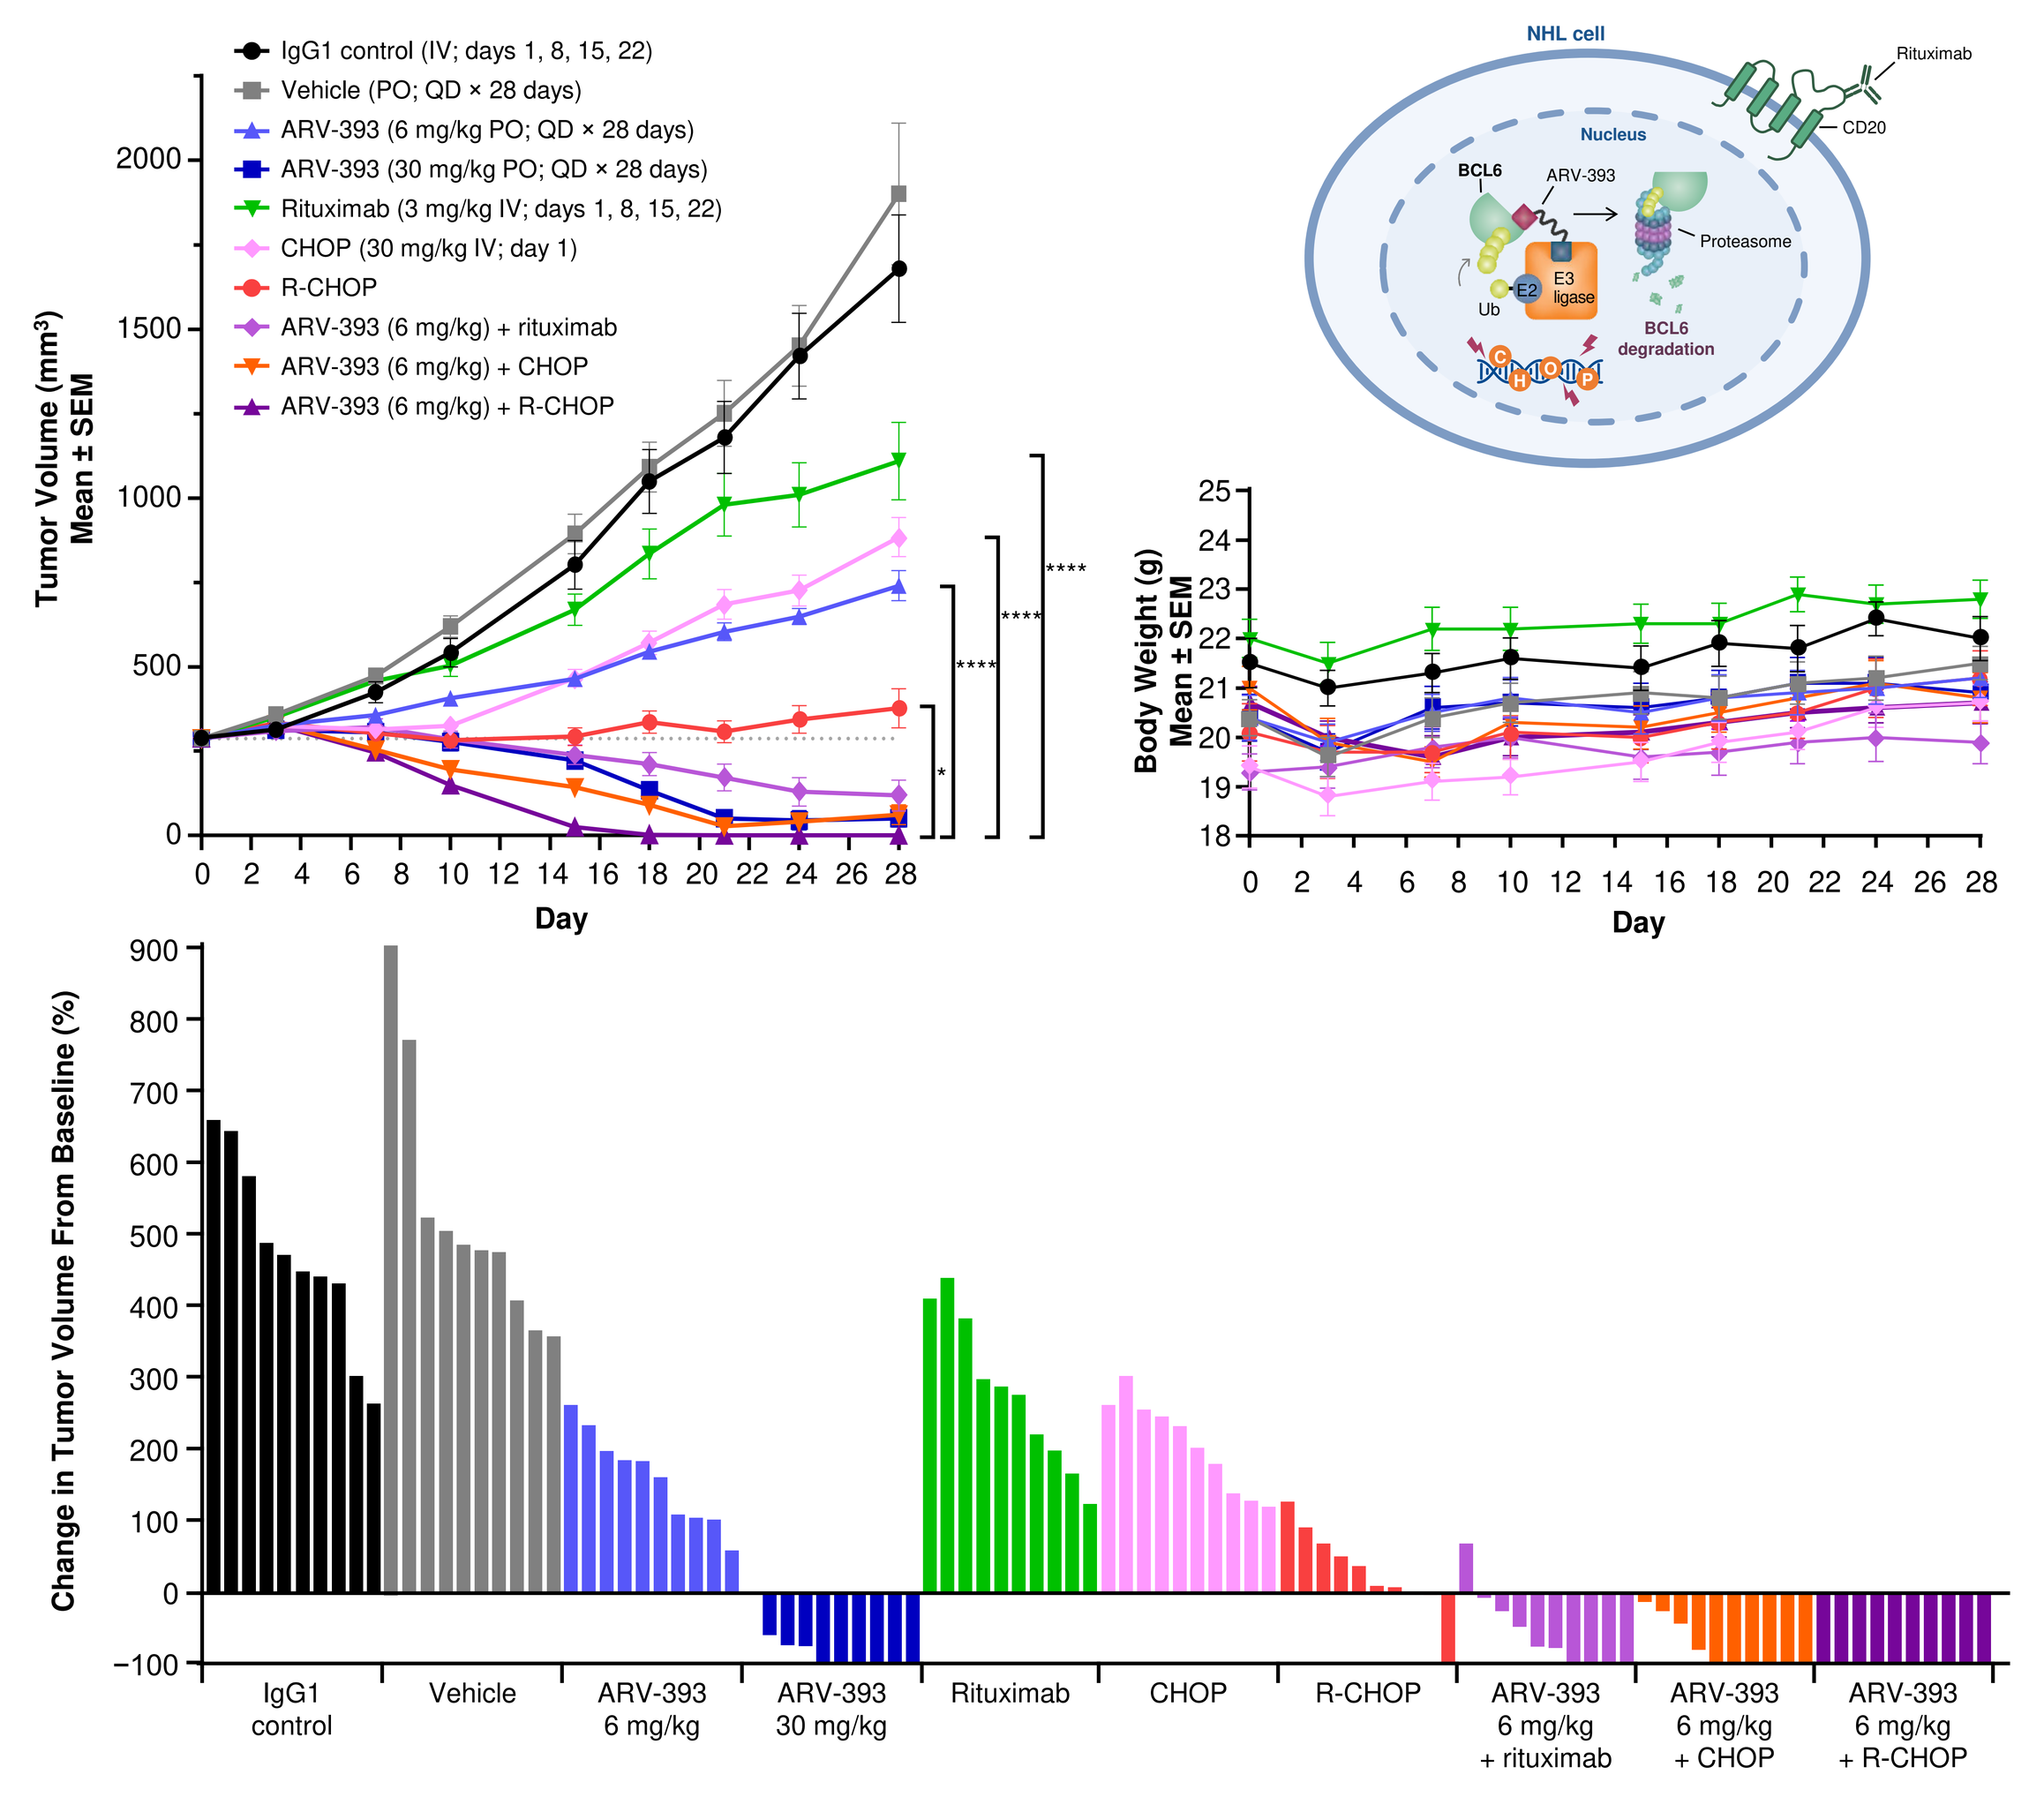
IgG1 control (IV; days 1, 8, 15, 22)
Vehicle (PO; QD × 28 days)
ARV-393 (6 mg/kg PO; QD × 28 days)
ARV-393 (30 mg/kg PO; QD × 28 days)
Rituximab (3 mg/kg IV; days 1, 8, 15, 22)
CHOP (30 mg/kg IV; day 1)
R-CHOP
ARV-393 (6 mg/kg) + rituximab
ARV-393 (6 mg/kg) + CHOP
ARV-393 (6 mg/kg) + R-CHOP
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
NHL cell
Rituximab
CD20
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation
Change in Tumor Volume From Baseline (%)
IgG1 control
Vehicle
ARV-393 6 mg/kg
ARV-393 30 mg/kg
Rituximab
CHOP
R-CHOP
ARV-393 6 mg/kg + rituximab
ARV-393 6 mg/kg + CHOP
ARV-393 6 mg/kg + R-CHOP

Mean tumor volume over time, animal body weight over time, and waterfall plot of individual tumor volume change from baseline to final measurement (day 28) for ARV-393 combined with rituximab, CHOP, or R-CHOP in a SU-DHL-4 HGBCL CDX mouse model. Body weight was measured twice weekly during the study.
$^{*}P<0.05$; $^{****}P<0.0001$ (one-way ANOVA, Tukey's multiple comparisons).
ANOVA=analysis of variance; BCL6=B-cell lymphoma 6; CD=cluster of differentiation; CDX=cell line–derived xenograft; CHOP=cyclophosphamide, hydroxydaunorubicin, vincristine sulfate, and prednisone; HGBCL=high-grade B-cell lymphoma; IgG1=immunoglobulin G1; IV=intravenously; NHL=non-Hodgkin lymphoma; PO=by mouth; QD=once daily; R-CHOP=rituximab and CHOP; SEM=standard error of the mean; Ub=ubiquitin.

Figure 4: ARV-393 in combination with (A) tafasitamab, (B) polatuzumab vedotin, or (C) rituximab

(A) Tafasitamab (SU-DHL-4 model)

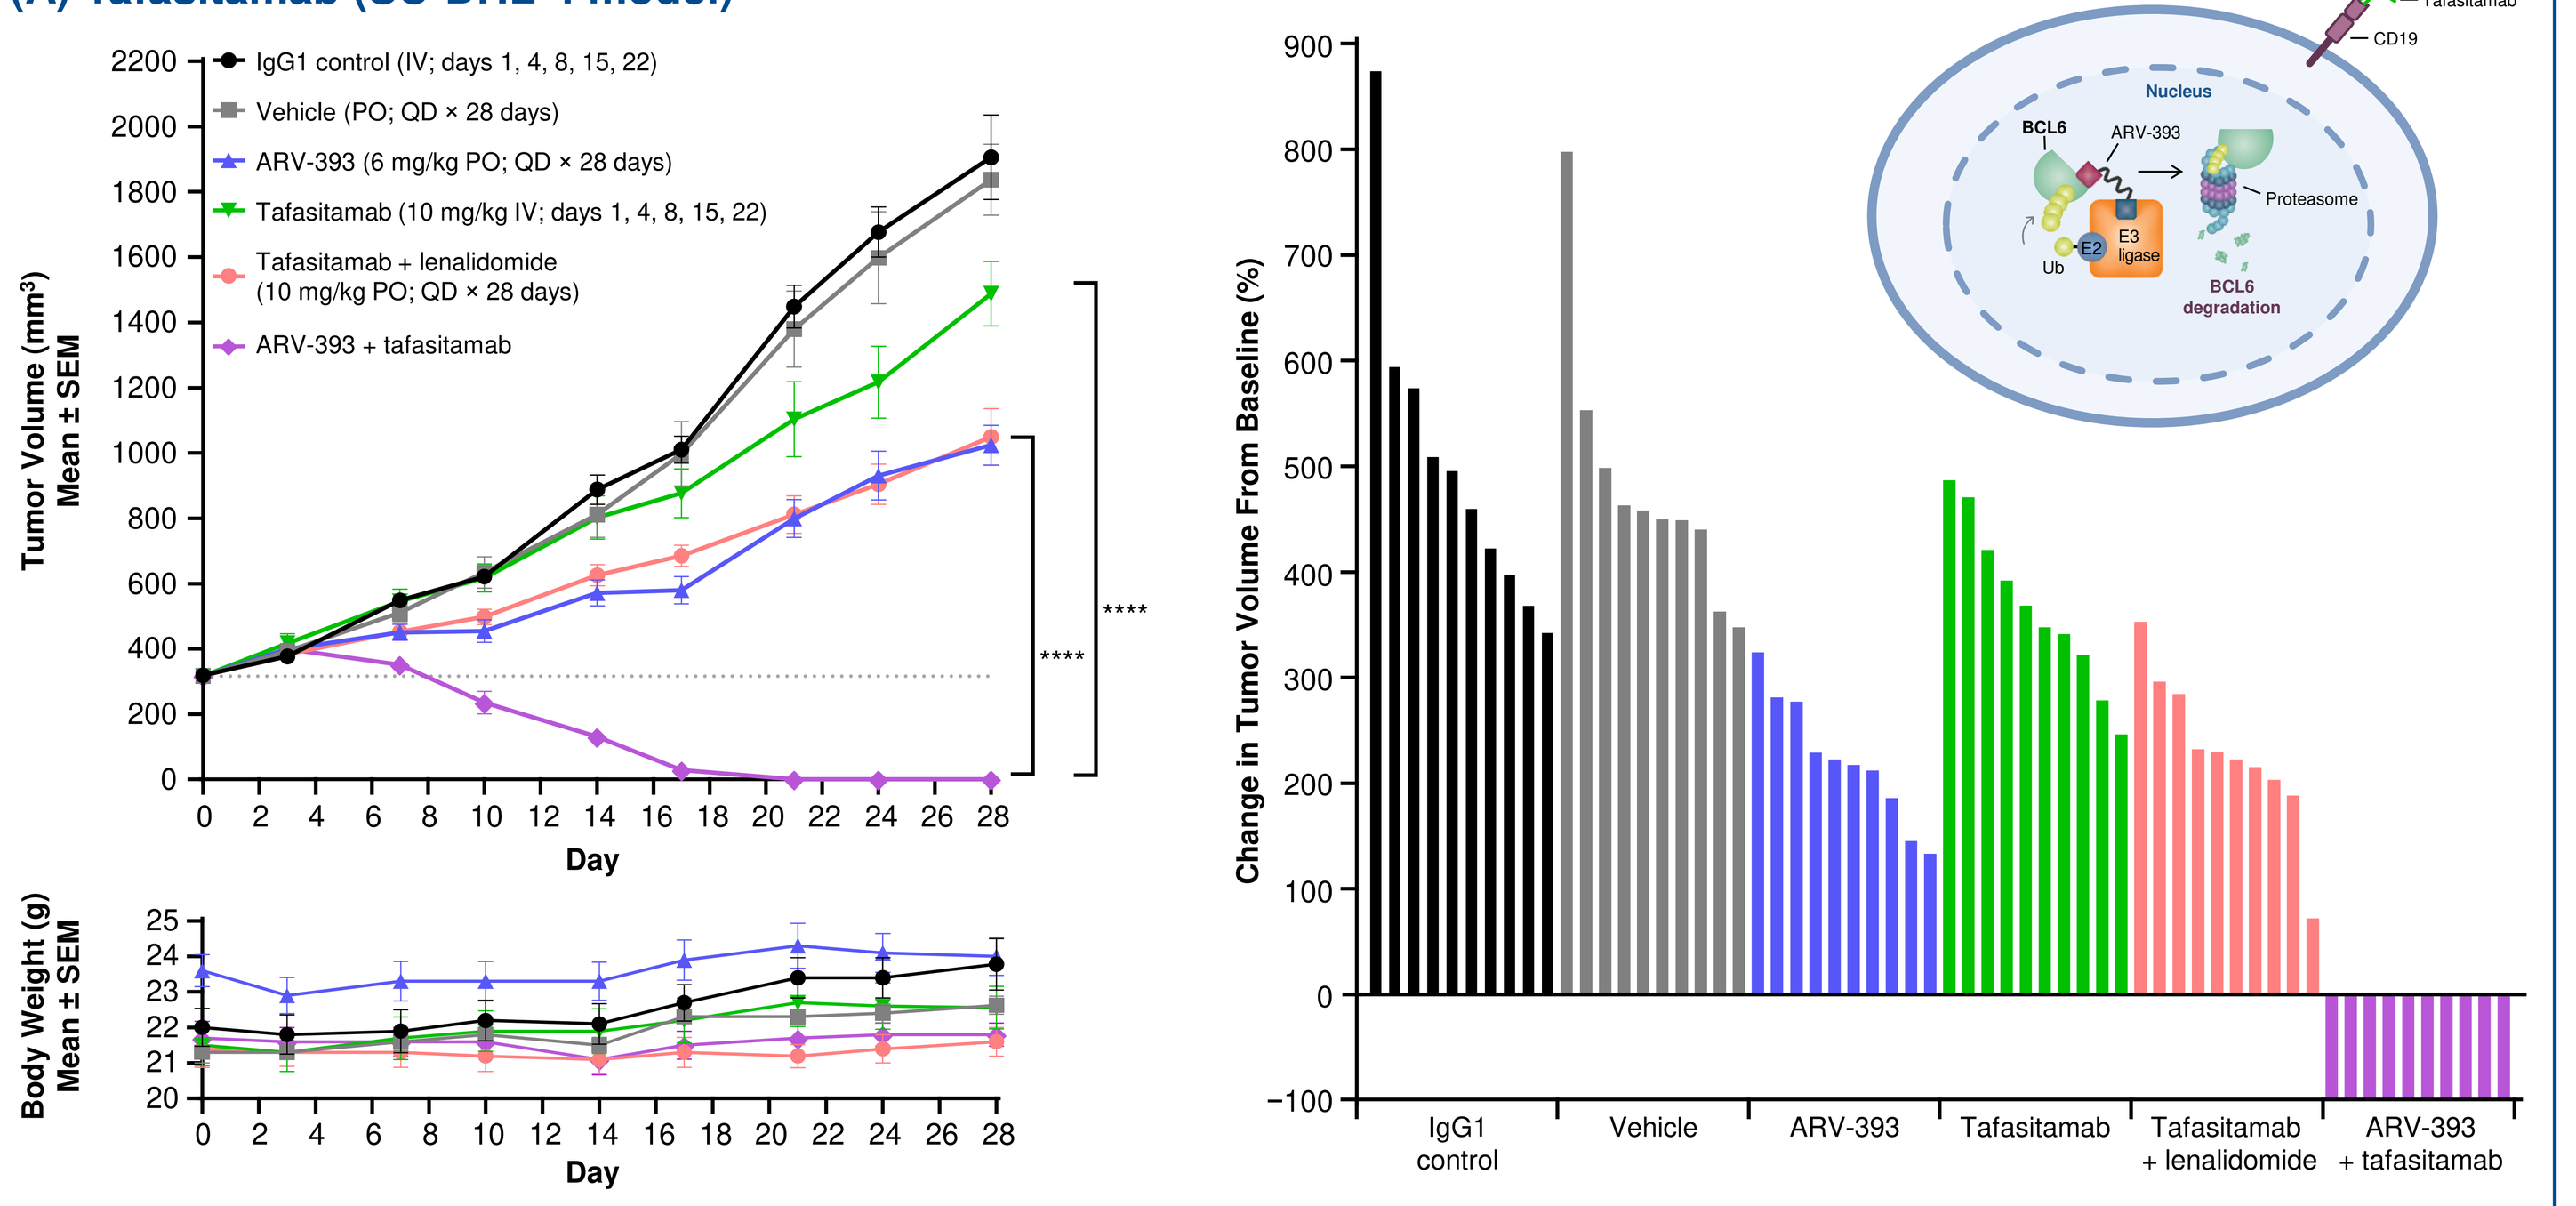
IgG1 control (IV; days 1, 4, 8, 15, 22)
Vehicle (PO; QD × 28 days)
ARV-393 (6 mg/kg PO; QD × 28 days)
Tafasitamab (10 mg/kg IV; days 1, 4, 8, 15, 22)
Tafasitamab + lenalidomide (10 mg/kg PO; QD × 28 days)
ARV-393 + tafasitamab
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
IgG1 control
Vehicle
ARV-393
Tafasitamab
Tafasitamab + lenalidomide
ARV-393 + tafasitamab
NHL cell
Tafasitamab
CD19
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

(B) Polatuzumab vedotin (SU-DHL-4 model)

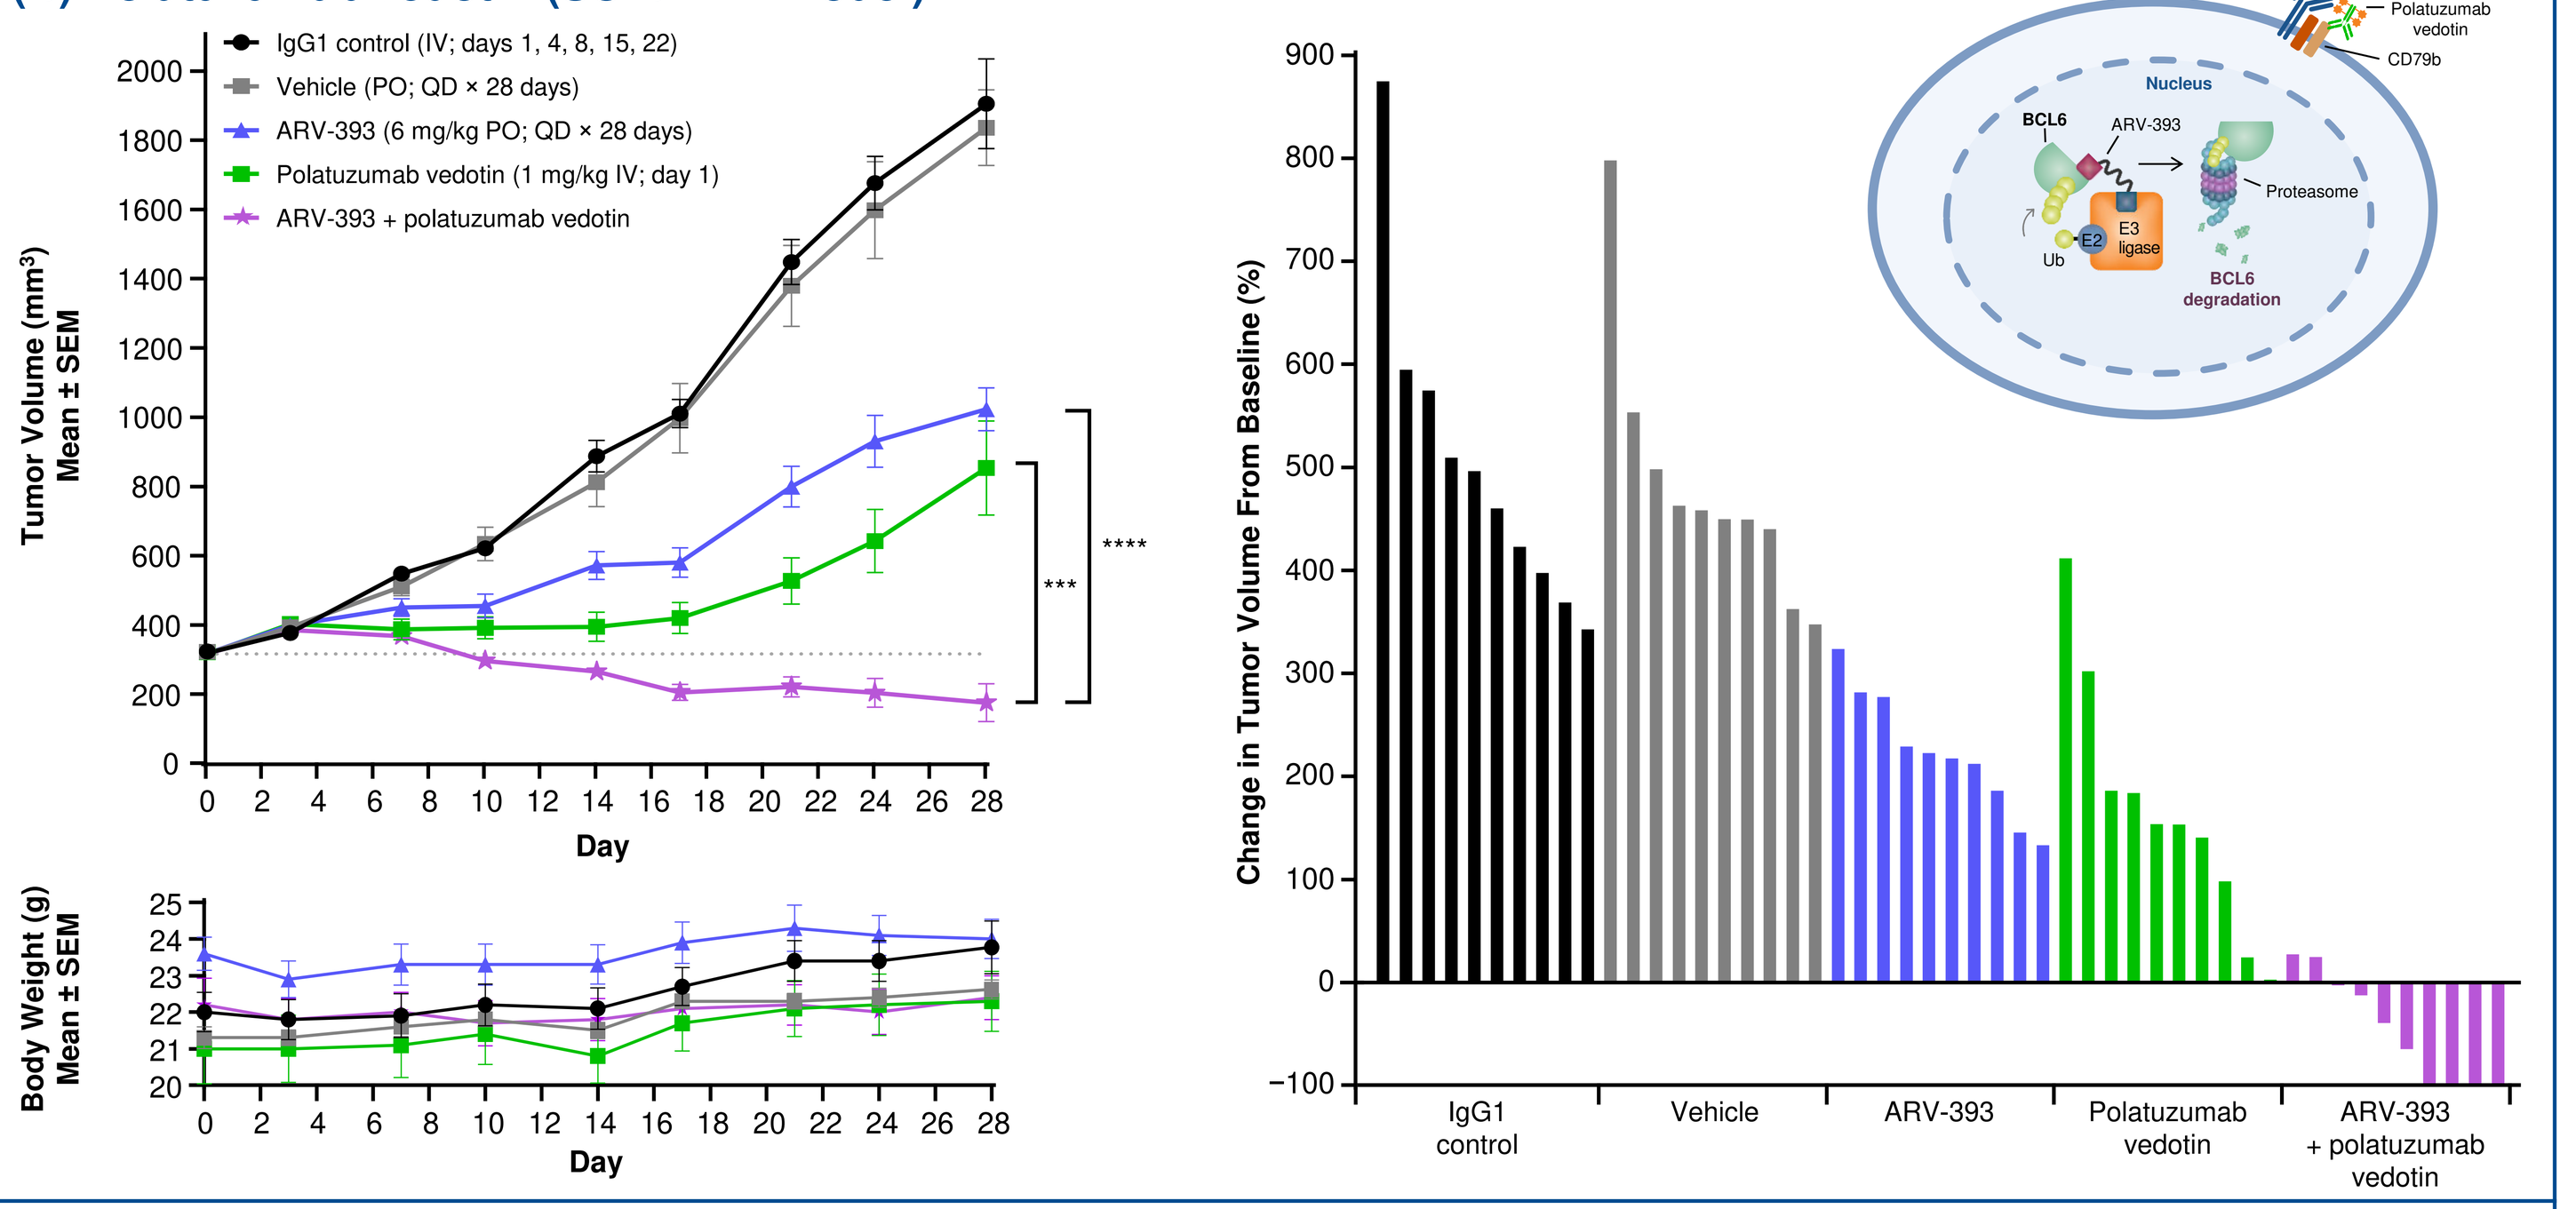
IgG1 control (IV; days 1, 4, 8, 15, 22)
Vehicle (PO; QD × 28 days)
ARV-393 (6 mg/kg PO; QD × 28 days)
Polatuzumab vedotin (1 mg/kg IV; day 1)
ARV-393 + polatuzumab vedotin
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
IgG1 control
Vehicle
ARV-393
Polatuzumab vedotin
ARV-393 + polatuzumab vedotin
NHL cell
BCR
Polatuzumab vedotin
CD79b
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

(C) Rituximab (SU-DHL-4 model)

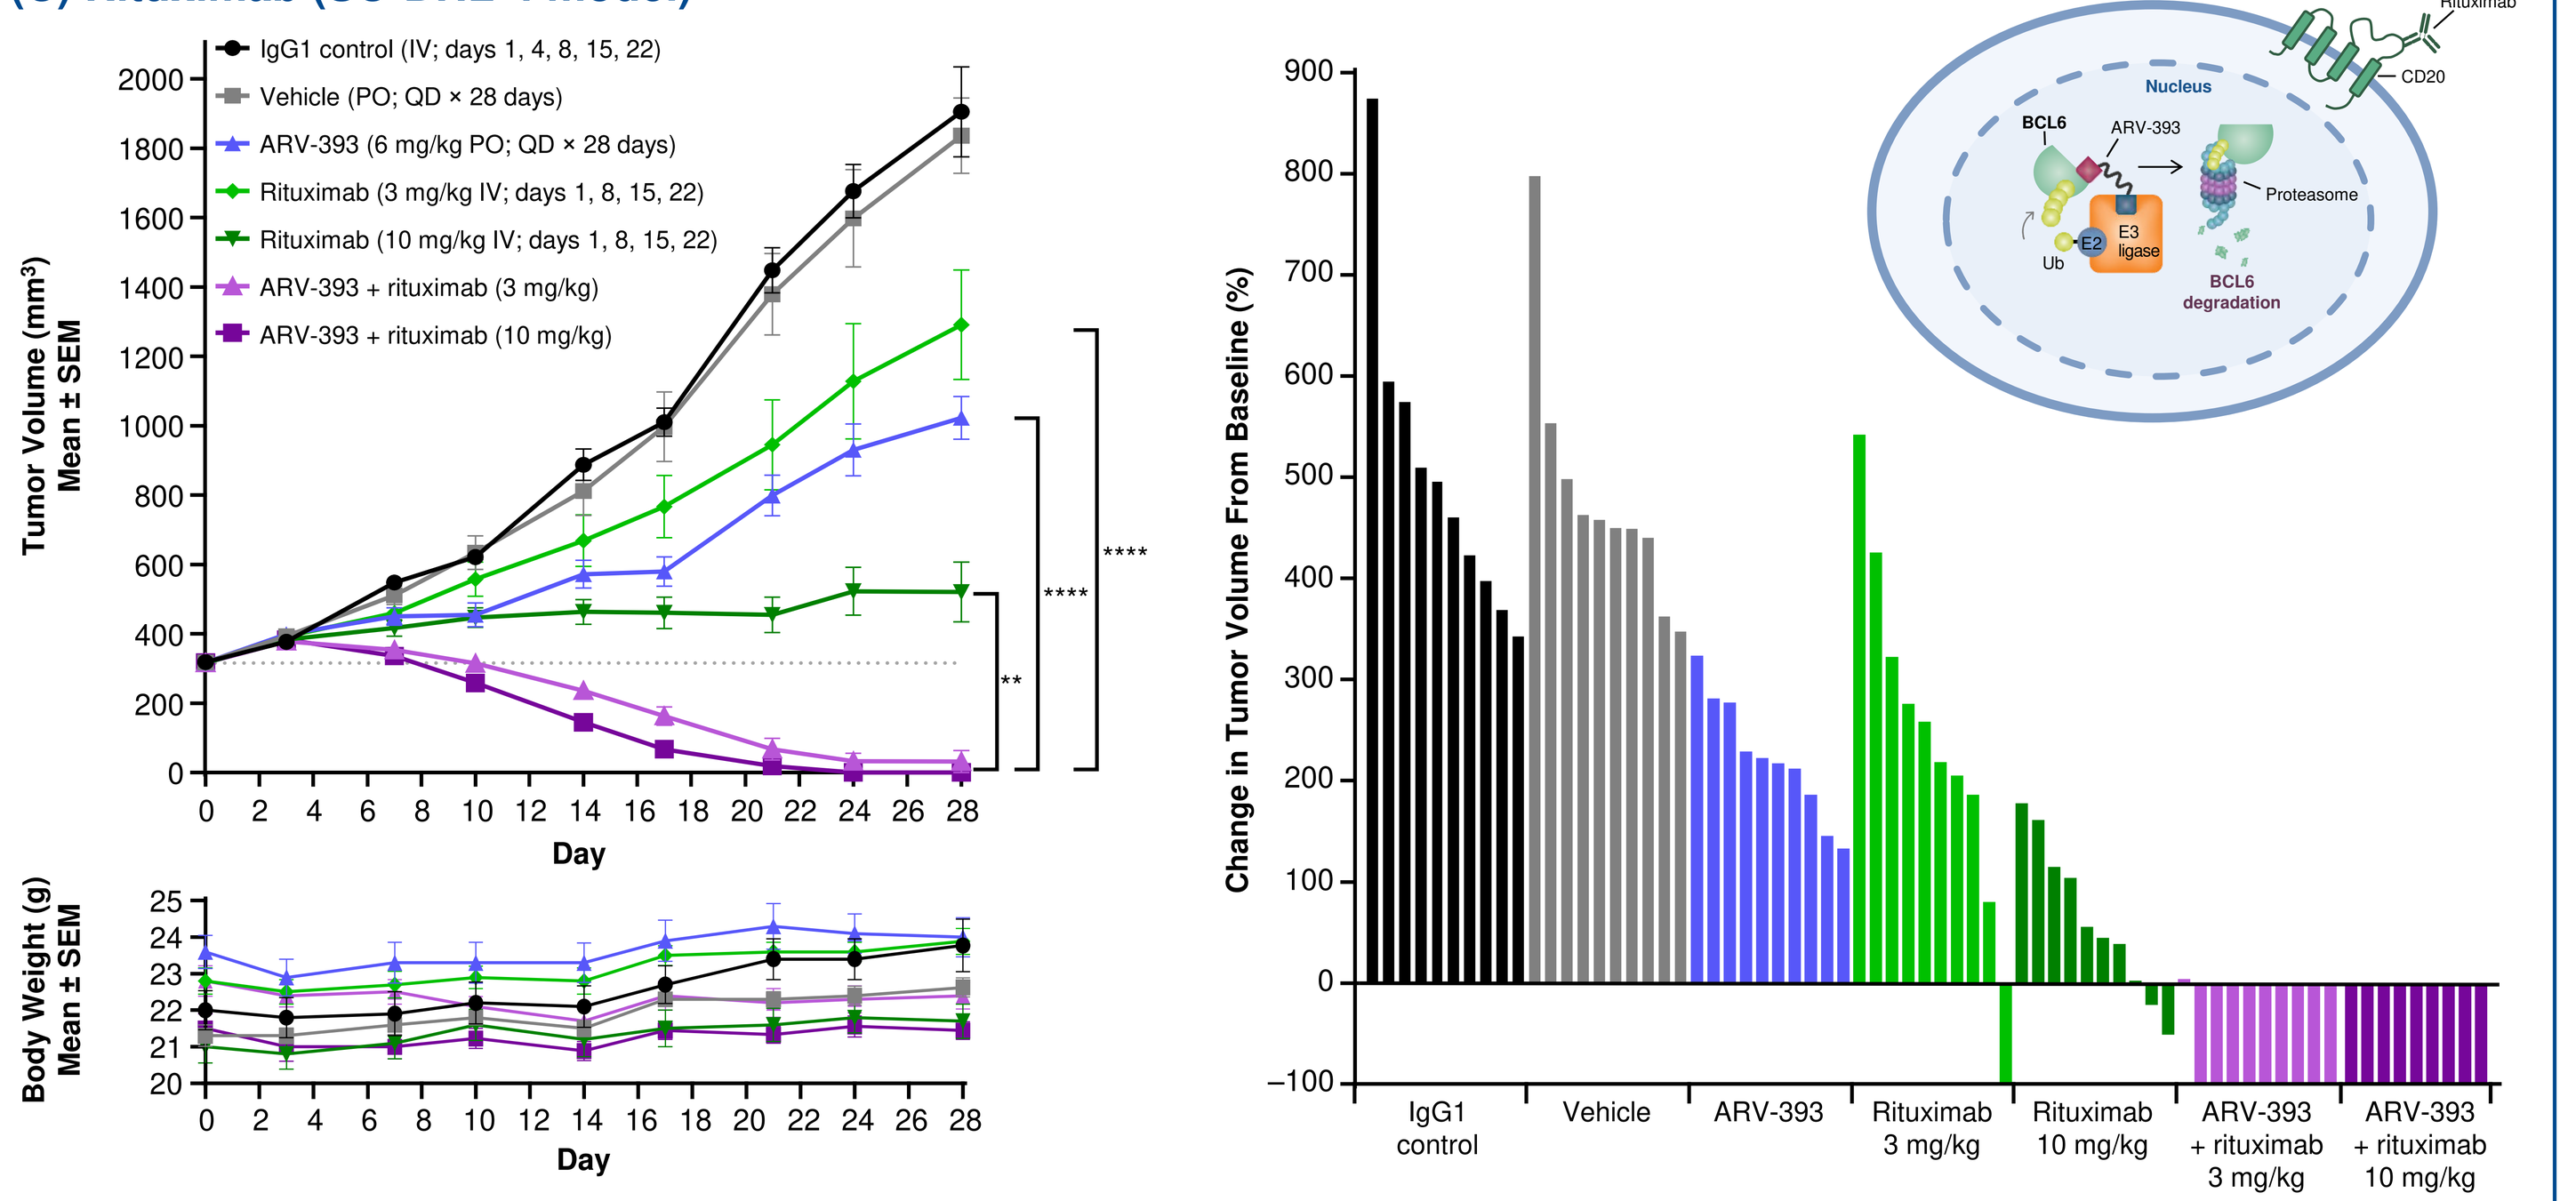
IgG1 control (IV; days 1, 4, 8, 15, 22)
Vehicle (PO; QD × 28 days)
ARV-393 (6 mg/kg PO; QD × 28 days)
Rituximab (3 mg/kg IV; days 1, 8, 15, 22)
Rituximab (10 mg/kg IV; days 1, 8, 15, 22)
ARV-393 + rituximab (3 mg/kg)
ARV-393 + rituximab (10 mg/kg)
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
IgG1 control
Vehicle
ARV-393
Rituximab 3 mg/kg
Rituximab 10 mg/kg
ARV-393 + rituximab 3 mg/kg
ARV-393 + rituximab 10 mg/kg
NHL cell
Rituximab
CD20
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

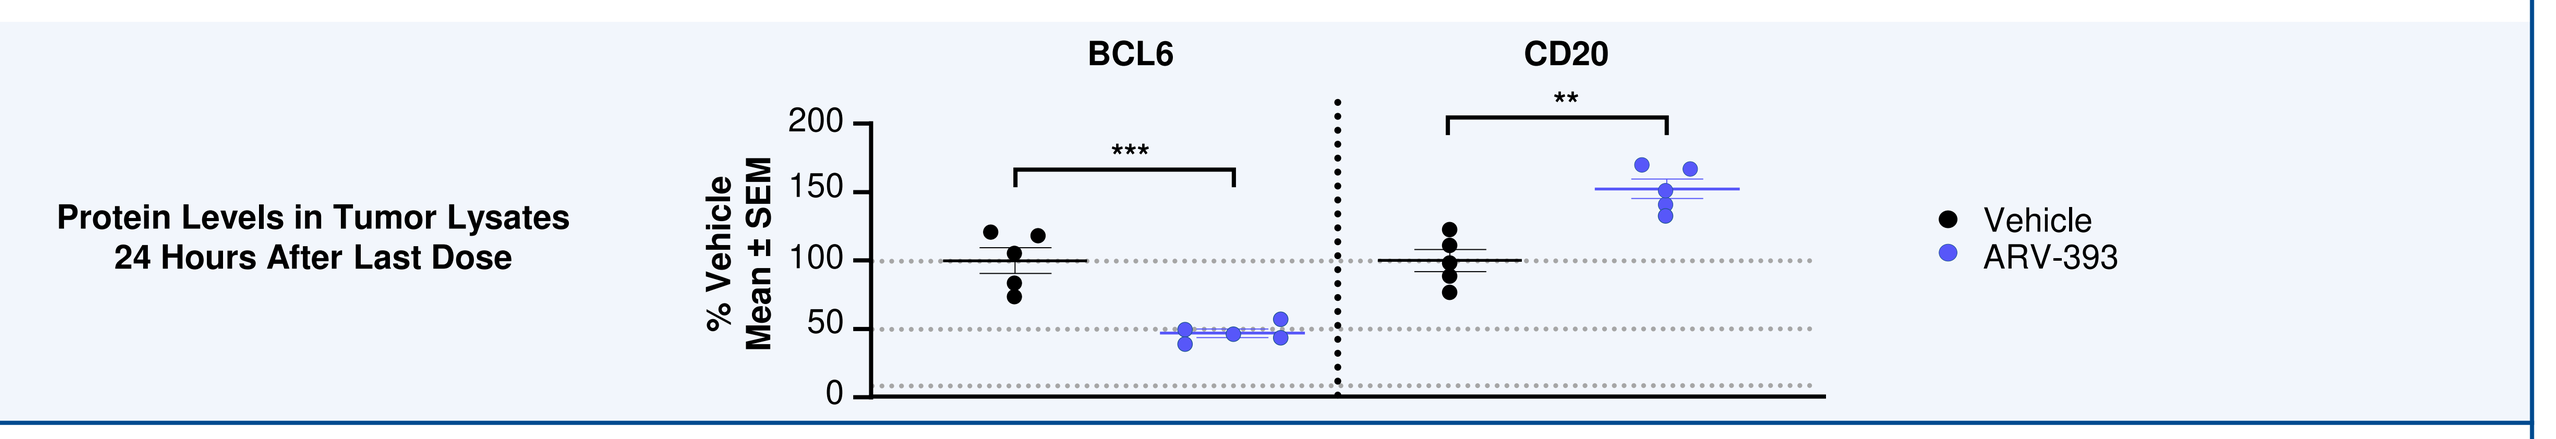
Protein Levels in Tumor Lysates
24 Hours After Last Dose
BCL6
CD20
% Vehicle Mean ± SEM
Vehicle
ARV-393

Mean tumor volume over time, animal body weight over time, and waterfall plot of individual tumor volume change from baseline to final measurement (day 28) for ARV-393 in combination with **(A)** tafasitamab (anti-CD19); **(B)** polatuzumab vedotin (anti-CD79b antibody-drug conjugate); or **(C)** rituximab (anti-CD20) in a SU-DHL-4 HGBCL CDX mouse model. Tumor lysate levels of BCL6 and CD20 proteins in vehicle- or ARV-393–treated mice 24 hours after the last dose are also shown in **(C)**. Body weight was measured twice weekly during the study. The same IgG1 IV control group, oral vehicle QD group, and ARV-393 monotherapy group were used in all panels.
$^{**}P<0.01$; $^{***}P<0.005$; $^{****}P<0.0001$ (one-way ANOVA, Tukey's multiple comparisons).
ANOVA=analysis of variance; BCL6=B-cell lymphoma 6; BCR=B-cell receptor; CD=cluster of differentiation; CDX=cell line–derived xenograft; HGBCL=high-grade B-cell lymphoma; IgG1=immunoglobulin G1; IV=intravenously; NHL=non-Hodgkin lymphoma; PO=by mouth; QD=once daily; SEM=standard error of the mean; Ub=ubiquitin.

Figure 5: ARV-393 in combination with (A) acalabrutinib, (B) venetoclax, or (C) tazemetostat

(A) Acalabrutinib (OCI-Ly10 model)

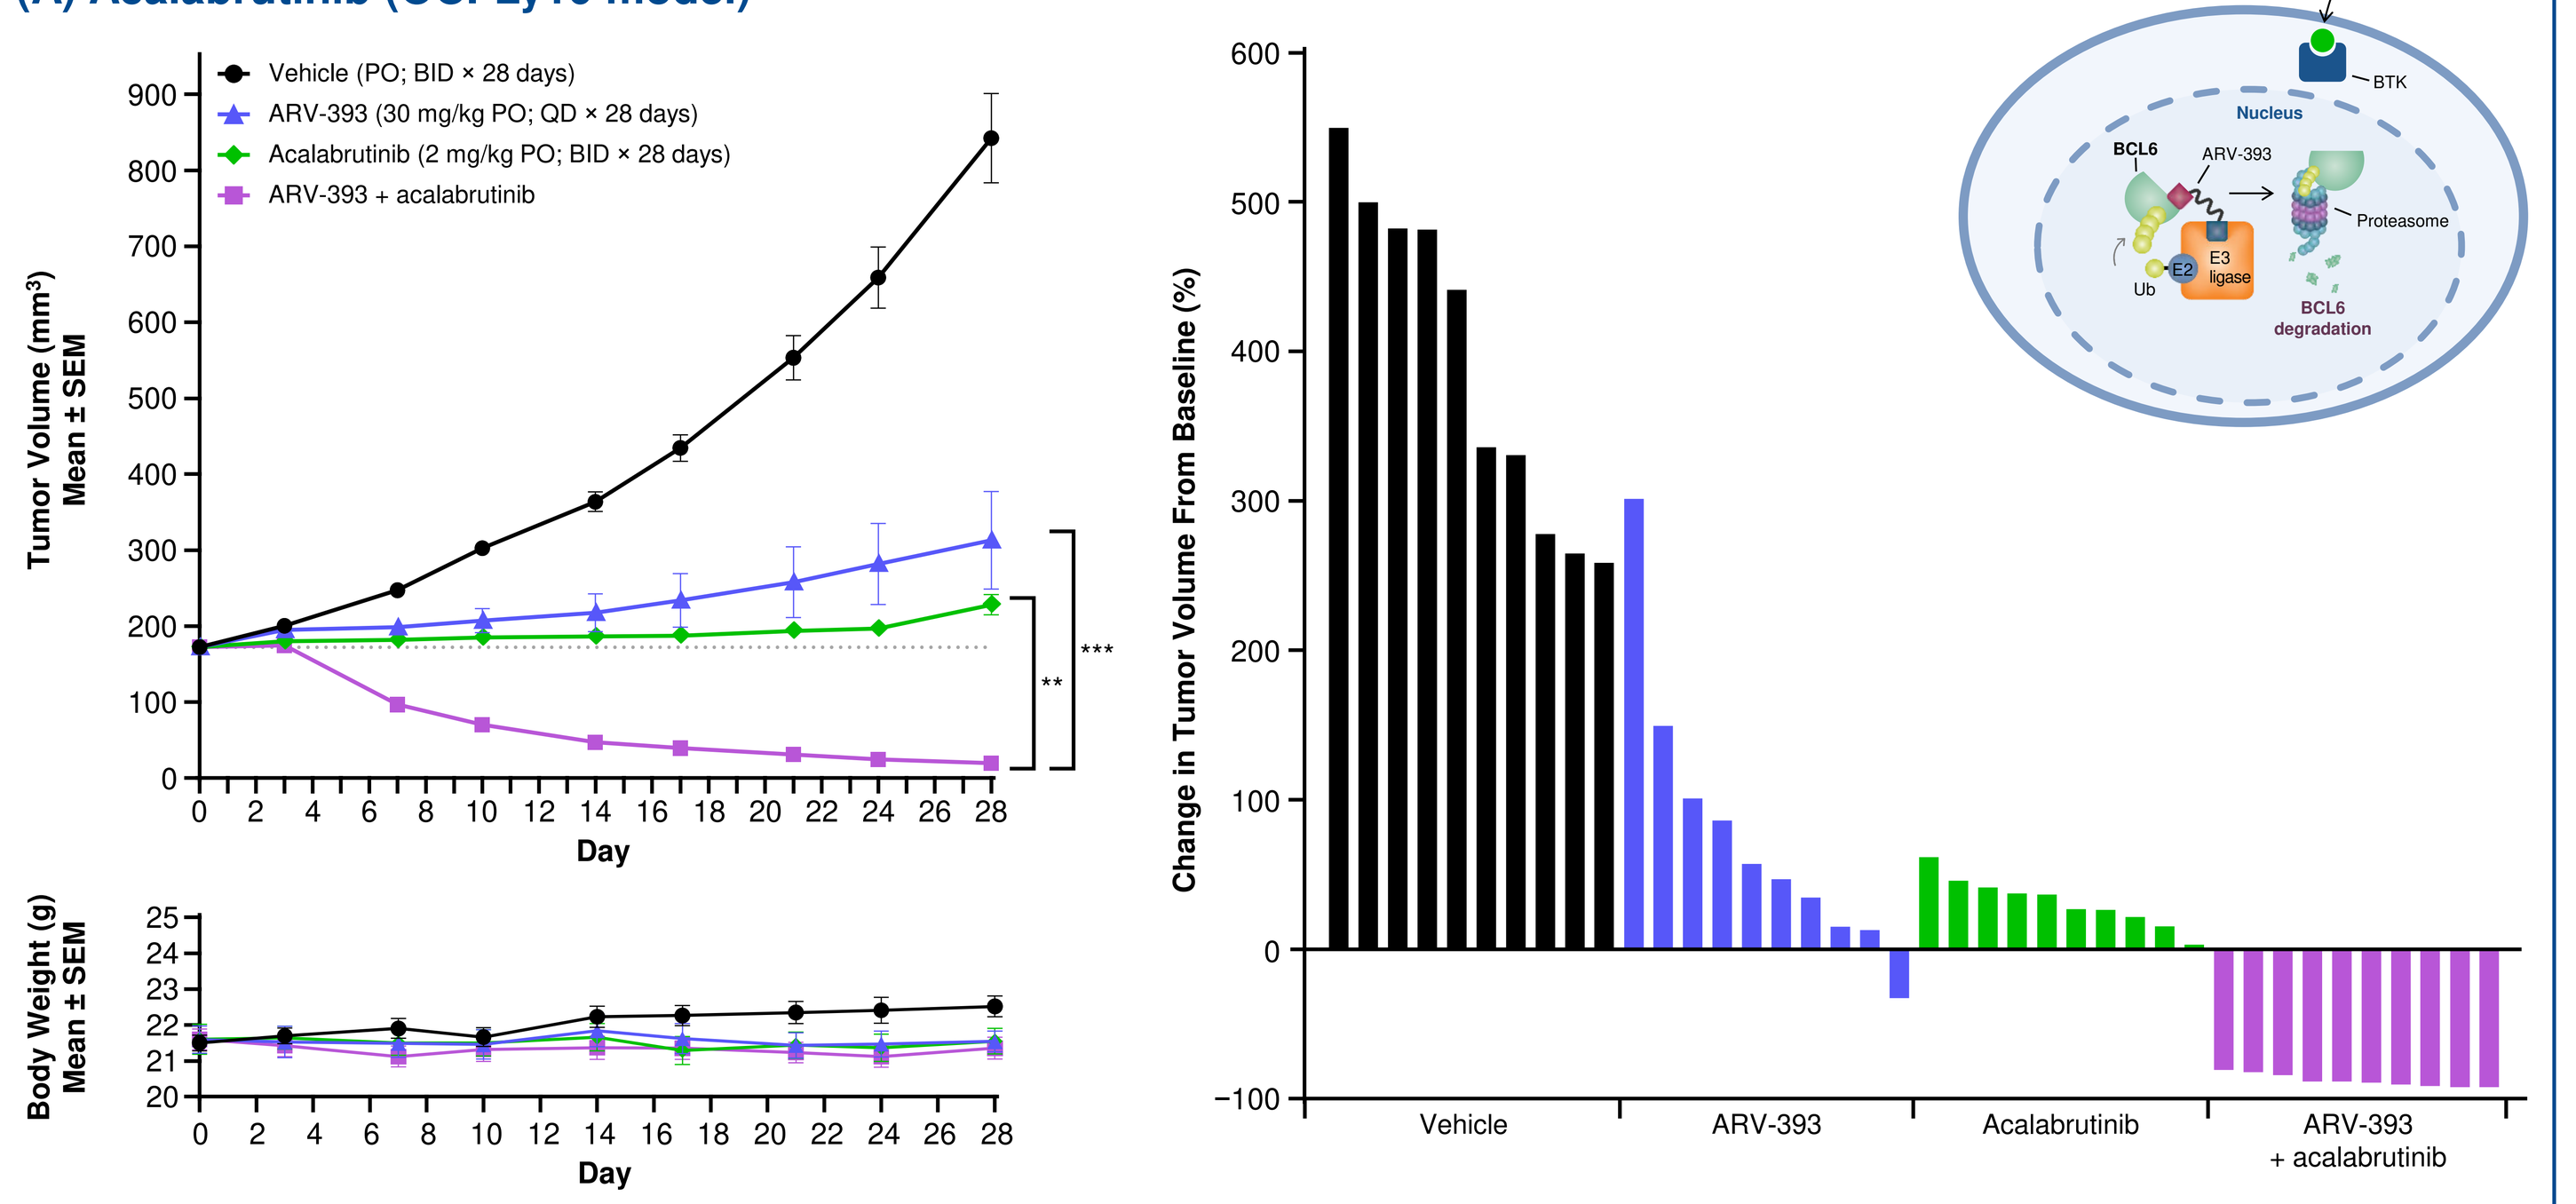
Vehicle (PO; BID × 28 days)
ARV-393 (30 mg/kg PO; QD × 28 days)
Acalabrutinib (2 mg/kg PO; BID × 28 days)
ARV-393 + acalabrutinib
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
Vehicle
ARV-393
Acalabrutinib
ARV-393 + acalabrutinib
NHL cell
Acalabrutinib
BTK
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

(B) Venetoclax (OCI-Ly1 model)

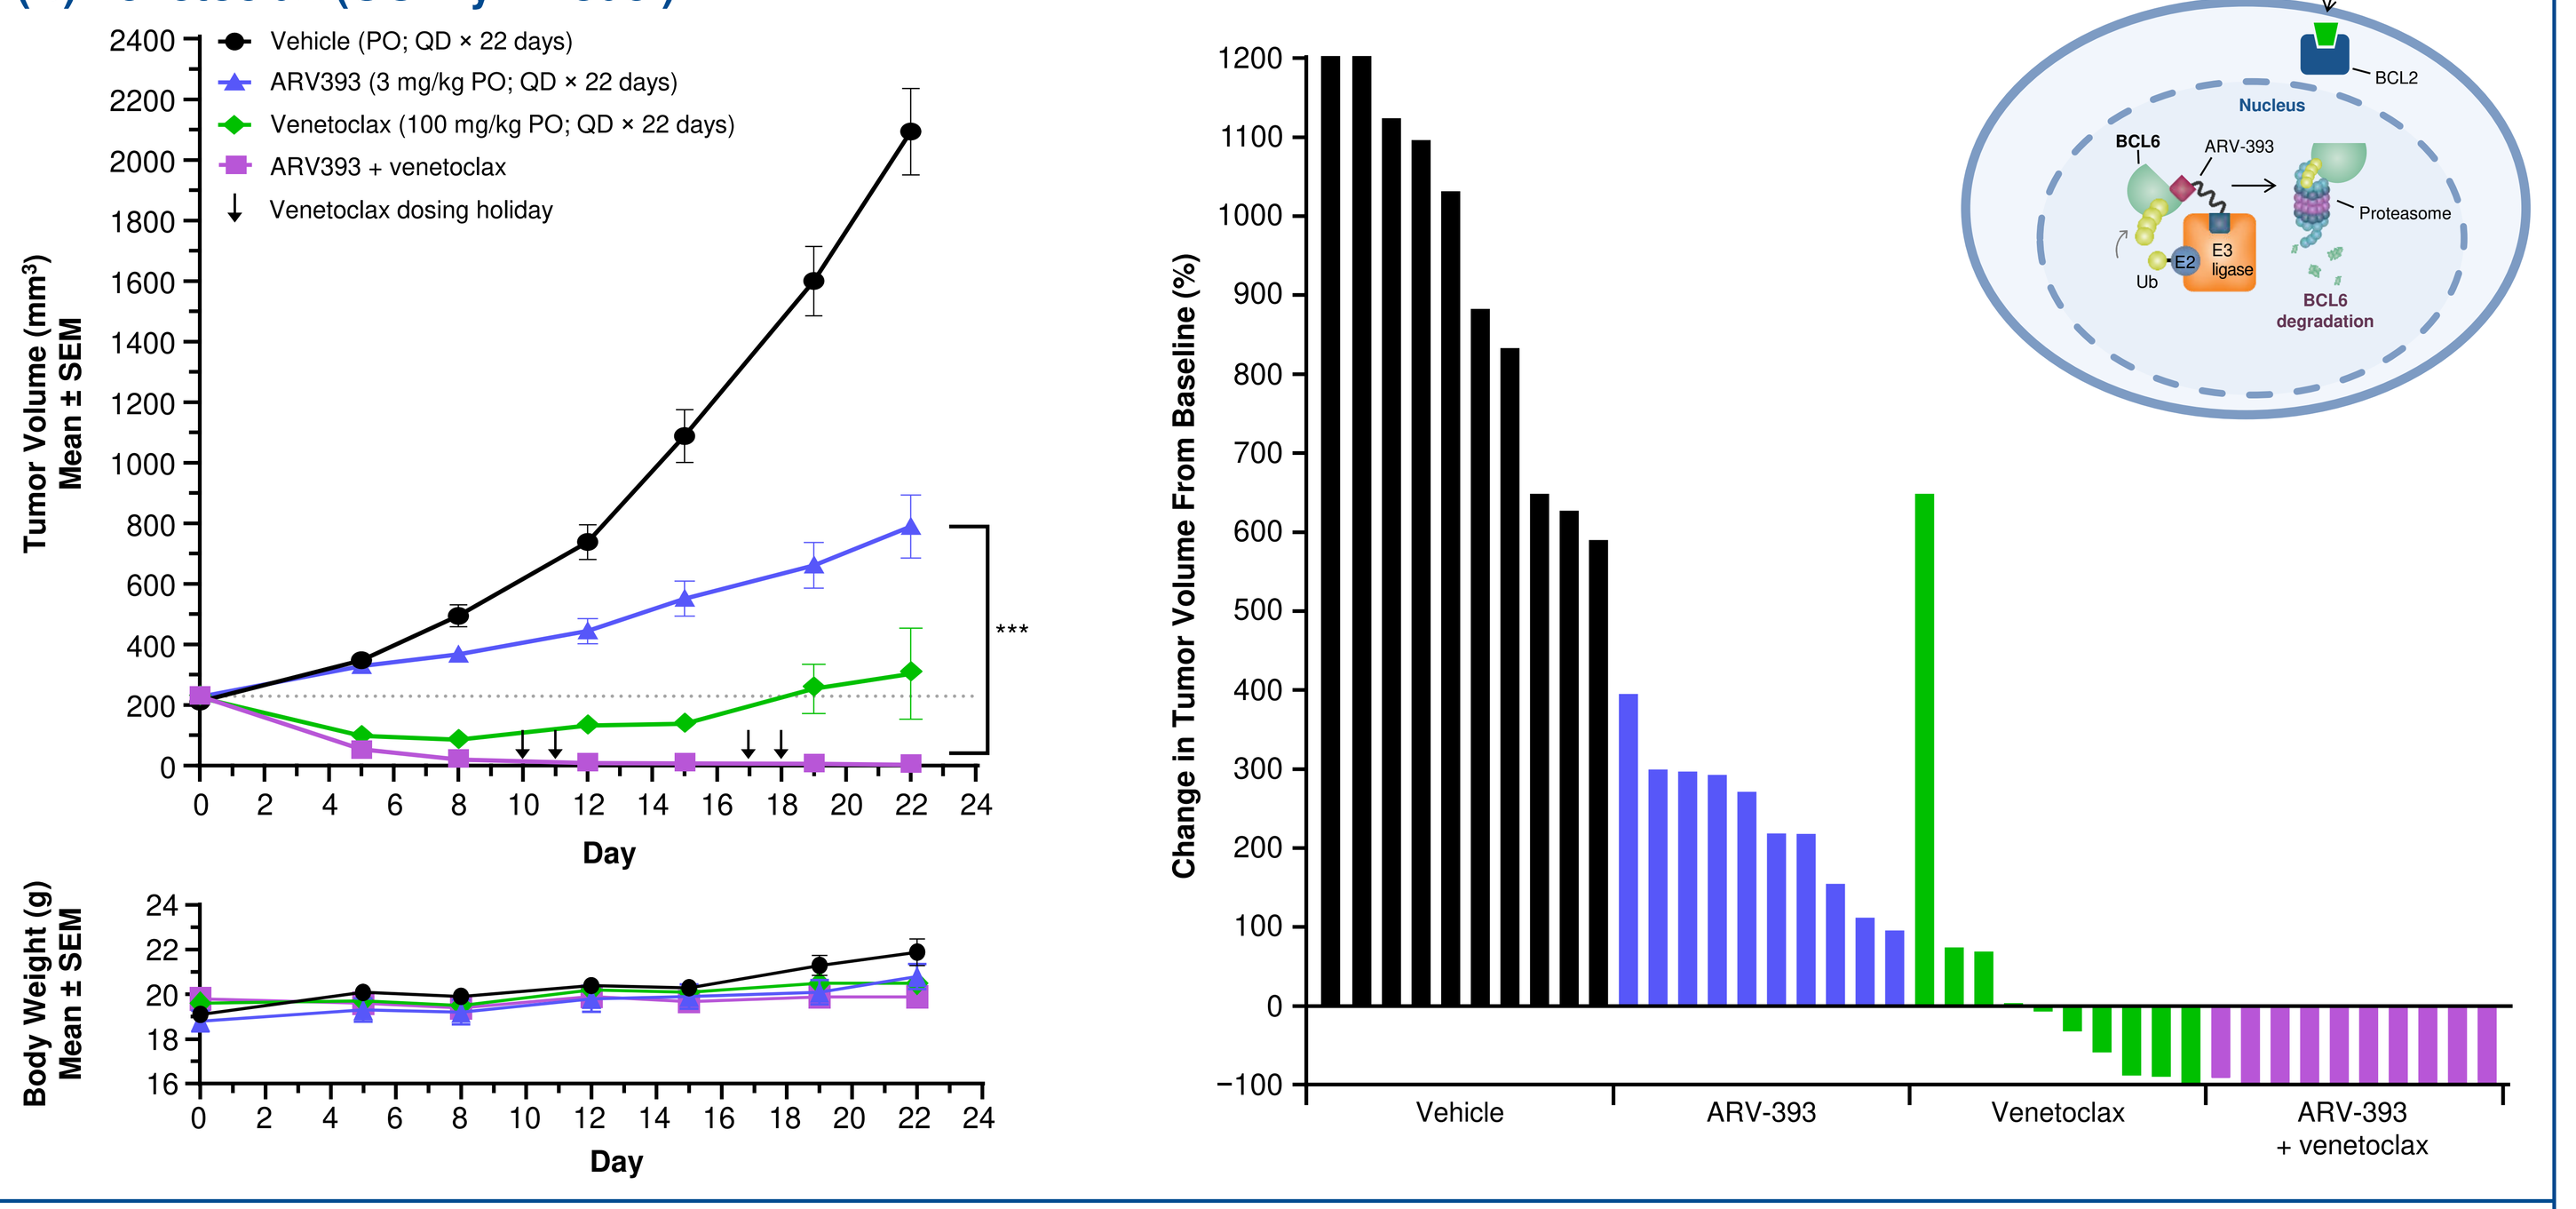
Vehicle (PO; QD × 22 days)
ARV393 (3 mg/kg PO; QD × 22 days)
Venetoclax (100 mg/kg PO; QD × 22 days)
ARV393 + venetoclax
Venetoclax dosing holiday
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
Vehicle
ARV-393
Venetoclax
ARV-393 + venetoclax
NHL cell
Venetoclax
BCL2
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

(C) Tazemetostat (SU-DHL-6 model)

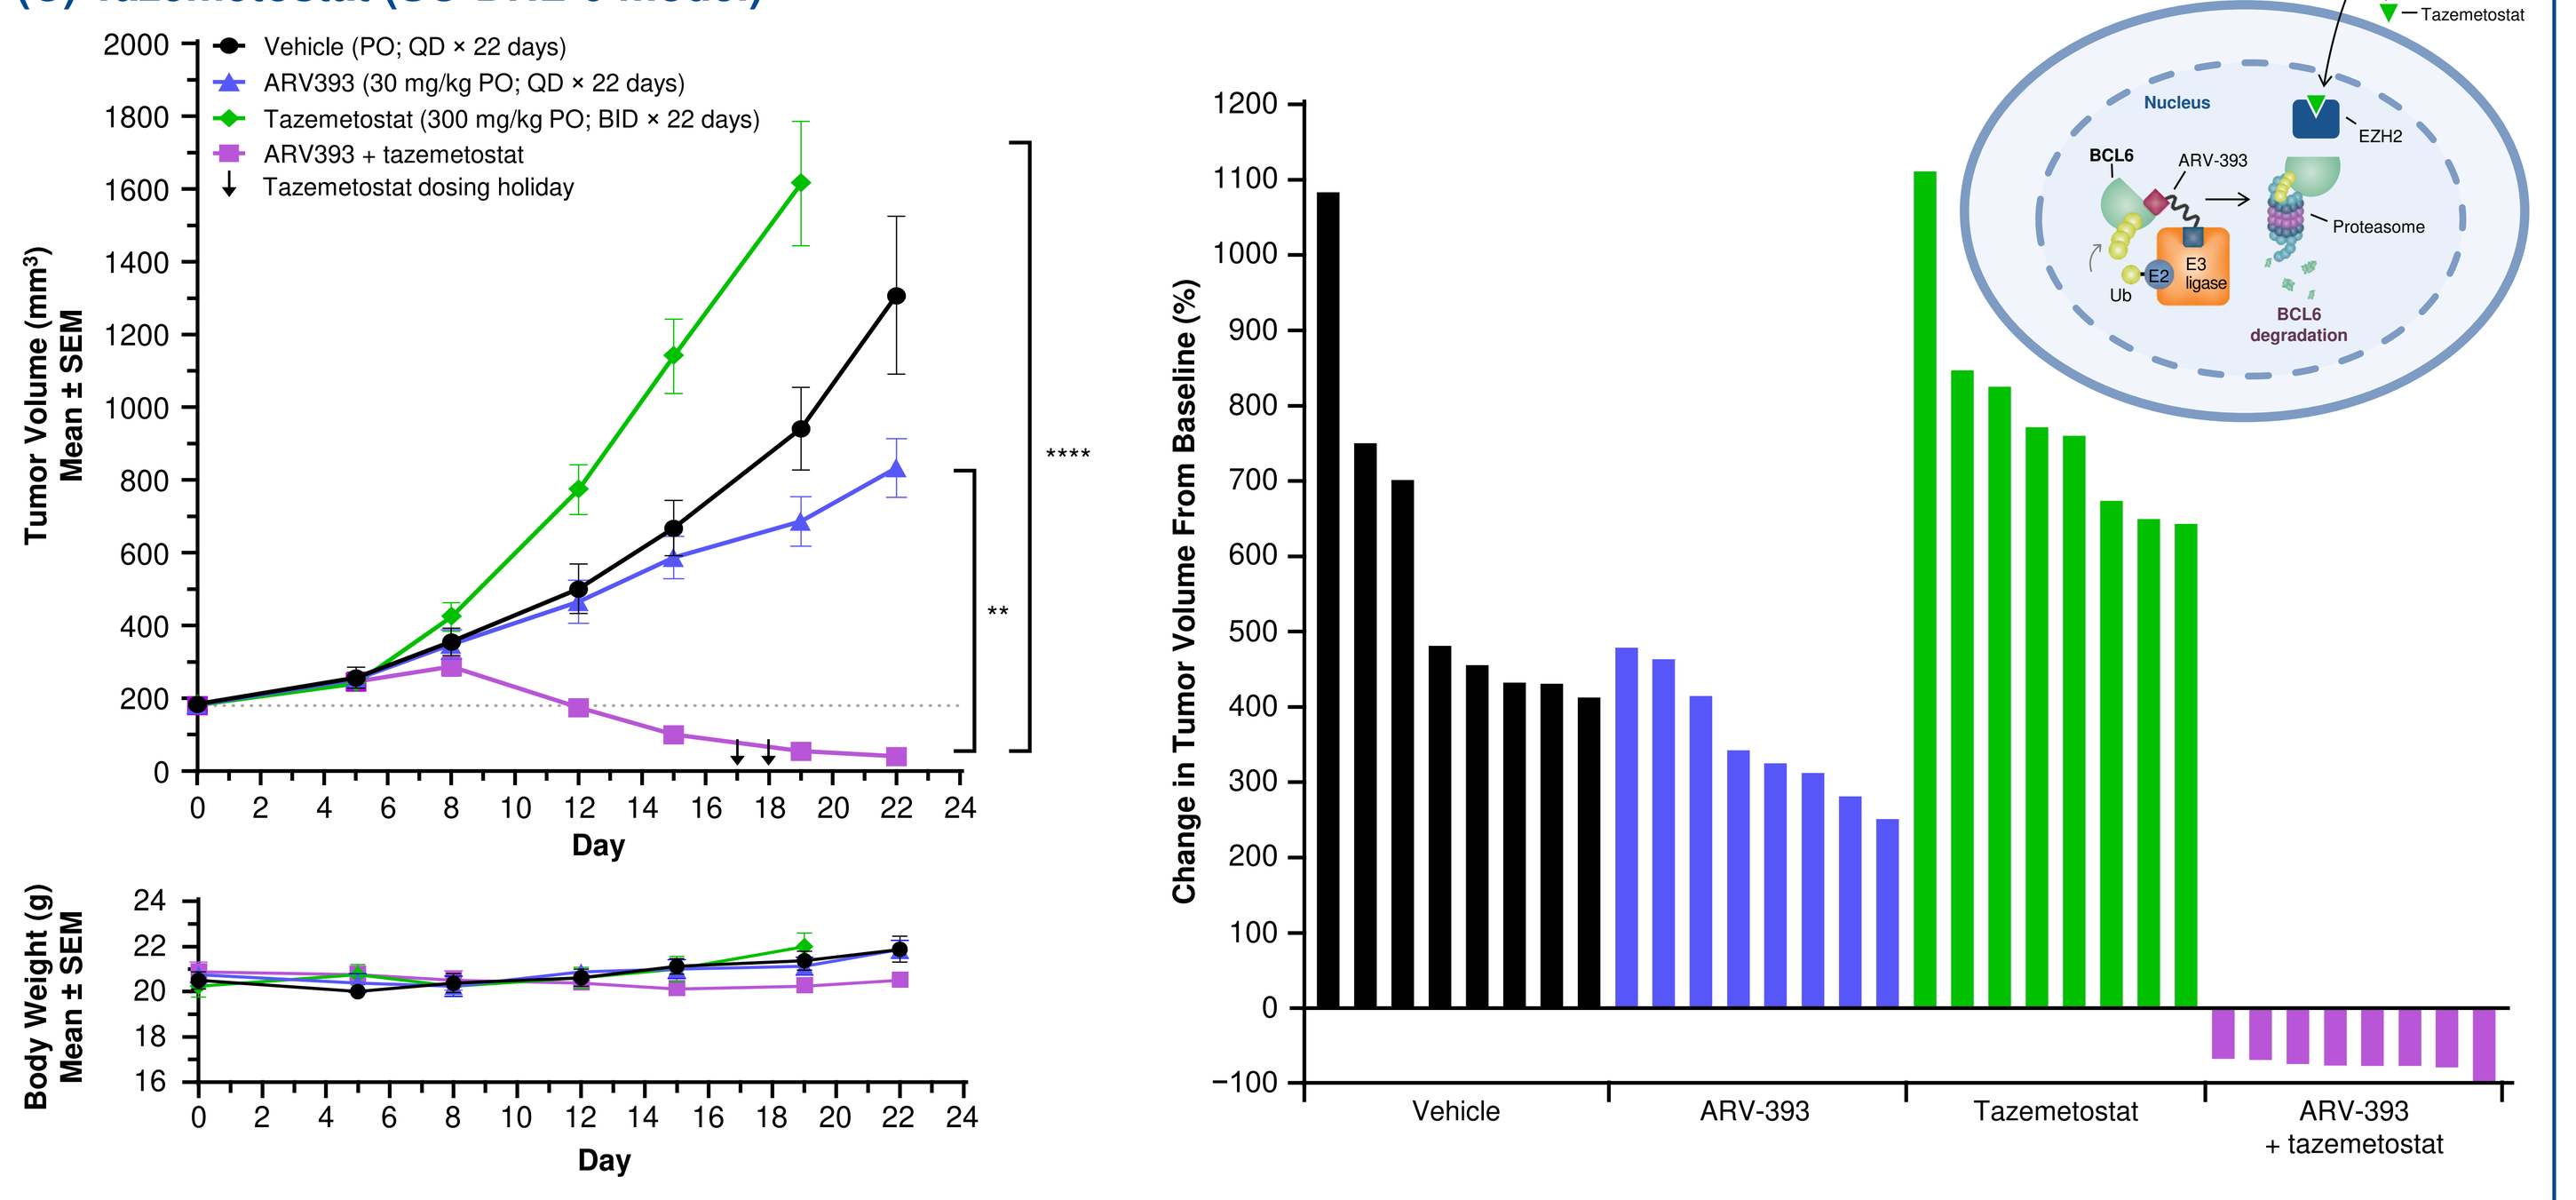
Vehicle (PO; QD × 22 days)
ARV393 (30 mg/kg PO; QD × 22 days)
Tazemetostat (300 mg/kg PO; BID × 22 days)
ARV393 + tazemetostat
Tazemetostat dosing holiday
Tumor Volume (mm³) Mean ± SEM
Body Weight (g) Mean ± SEM
Day
Change in Tumor Volume From Baseline (%)
Vehicle
ARV-393
Tazemetostat
ARV-393 + tazemetostat
NHL cell
Tazemetostat
EZH2
Nucleus
BCL6
ARV-393
Proteasome
E2
E3 ligase
Ub
BCL6 degradation

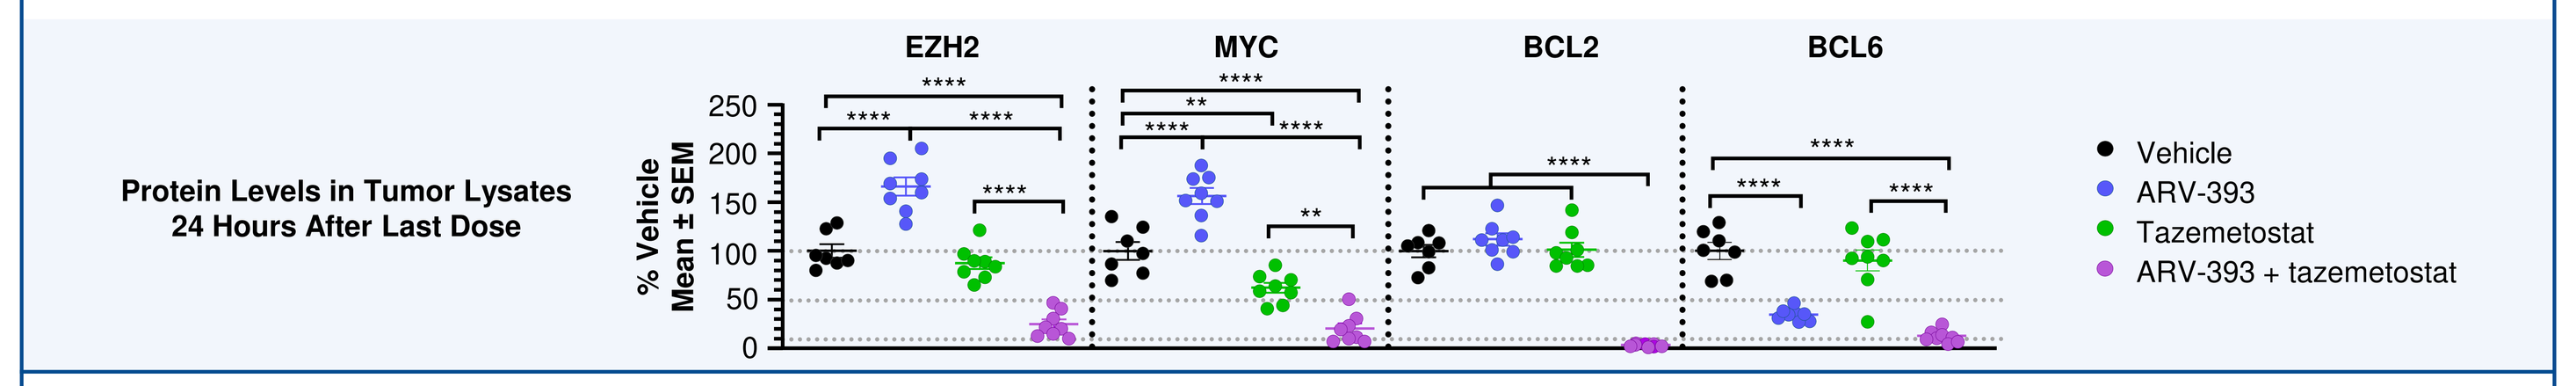
Protein Levels in Tumor Lysates
24 Hours After Last Dose
EZH2
MYC
BCL2
BCL6
% Vehicle Mean ± SEM
Vehicle
ARV-393
Tazemetostat
ARV-393 + tazemetostat

Mean tumor volume over time, animal body weight over time, and waterfall plot of individual tumor volume change from baseline to final measurement (day 28 or 22) for ARV-393 in combination with **(A)** acalabrutinib (BTK SMI) in an ABC OCI-Ly10 (*MYD88*-mutant) CDX mouse model; **(B)** venetoclax (BCL2 SMI) in a BCL2+ OCI-Ly1 CDX mouse model; or **(C)** tazemetostat (EZH2 SMI) in an *EZH2*-mutant SU-DHL-6 CDX mouse model. Tumor lysate levels of EZH2, MYC, BCL2, and BCL6 proteins in vehicle-, ARV-393–, tazemetostat-, or ARV-393 + tazemetostat–treated mice 24 hours after the last dose are also shown in **(C)**. Body weight was measured twice weekly during the study.
$^{**}P<0.01$; $^{***}P<0.005$; $^{****}P<0.0001$ (one-way ANOVA, Tukey's multiple comparisons).
ABC=activated B-cell; ANOVA=analysis of variance; BCL2=B-cell lymphoma 2; BCL6=B-cell lymphoma 6; BID=twice daily; BTK= Bruton tyrosine kinase; CDX=cell line–derived xenograft; EZH2=enhancer of zeste homolog 2; NHL=non-Hodgkin lymphoma; PO=by mouth; QD=once daily; SEM=standard error of the mean; SMI=small-molecule inhibitor; Ub=ubiquitin.