Mechanisms of Acquired Resistance to Vepdegestrant (ARV-471), a Novel PROTAC Estrogen Receptor (ER) Degrader

Jessica Teh, Shannon Bessonett, Wendy Wu, Christopher Kuhlberg, Alissa Wynne, Sean Landrette, Monica Andreoli, Elizabeth Bortolon, Jennifer Pizzano, Richard Gedrich and Ian Taylor

Arvinas Estrogen Receptor, Inc., New Haven, CT

Background

Vepdegestrant (ARV-471) is an orally bioavailable cereblon (CRBN)-based PROteolysis-TArgeting Chimera (PROTAC®) small molecule that demonstrates superior ER degradation and anti-tumor activity compared to fulvestrant in endocrine sensitive and resistant xenograft models (1,2) and has shown significant ER degradation and promising clinical benefit in late-line ER-positive breast cancer patients (3,4). Innate and acquired drug resistance limits the response to and durability of many cancer therapies.

Objective

- The purpose of this study is to identify potential mechanisms of resistance to ARV-471, an oral ER PROteolysis TArgeting Chimera (PROTAC®) protein degrader.
- Characterization of ARV-471 resistant MCF7 cell lines were conducted using Whole Exome Sequencing (WES), RNA sequencing (RNAseq) and Reverse Phase Protein Array (RPPA).

Key Findings

- Acquired resistance to ARV-471 was associated with downregulation of ER protein expression and signaling and upregulation of HER family (EGFR, HER2, HER3) and MAPK/AKT signaling.
- Whole exome sequencing identified an *NRAS* copy number gain in one ARV-471-resistant cell line. No genomic alterations in *ESR1*, *CRBN* or HER family members were observed.
- NRAS and EGFR overexpression in MCF7 cells conferred resistance to ARV-471 *in vitro*.
- ARV-471-resistant cells with HER pathway upregulation were sensitive to EGFR, pan-HER, MEK and ERK inhibitors.
- CRBN knockout in ER+ breast cancer cells prevented ER degradation, but did not confer resistance to ARV-471, consistent with ARV-471 possessing ER antagonist activity independent of its degrader activity.

Conclusions

- Together, these data suggest that acquired resistance to ARV-471 may be associated with alterations within Receptor Tyrosine Kinase/MAPK signaling pathways rather than ER signaling or E3 ligase machinery.

References

1. Flanagan J et al. *Cancer Res.* 2019;79 (4 Supplement): P5-04-18
2. Snyder L et al. *Cancer Res.* 2021;81 (13 Supplement): 44
3. Hamilton E et al. *Cancer Res.* 2022;82 (4 Supplement): PD13-08
4. Hurvitz SA et al. SABCS 2022
5. Farshidfar F et al. *Nat. Commun.* 2022;13(1):898

Contact

Jessica Teh; jessica.teh@arvinas.com

Presented at the American Association for Cancer Research Annual Meeting, Orlando FL 2023

Methods

- **Generation of MCF7 cell lines with acquired resistance to ARV-471**
 - To generate resistant cells *in vitro*, MCF7 cells were cultured with increasing concentrations of ARV-471 up to 1 µM for approximately 8 weeks. Single cell clones (labeled 3B3 and 4A8) were then isolated.
 - For *in vivo* generation of resistant cells, 471R1 cells were isolated from an MCF7 xenograft tumor that was treated with once daily ARV-471 at 3 mg/kg for 53 days. 471R2 cells were isolated from an MCF7 xenograft tumor that was treated with once daily ARV-471 at 1 mg/kg for 30 days followed by 3 mg/kg of ARV-471 for 10 days.
 - All resistant cell lines were maintained in culture at 1 µM ARV-471.
- **Whole exome sequencing (WES)**
 - Sequencing was performed at Yale Center for Genome Analysis (YCGA) on the Novaseq targeting 150x for resistant cells and 80x for parental cells. The sequence data was analyzed with a combination of germline and somatic variant calling to identify somatic variants, candidate loss-of-heterozygosity (LOH), as well as copy number variation (CNV) events. The detailed procedures and filtering criteria are described in the publication (5).
- **RNAseq**
 - Resistant cells were continuously cultured in ARV-471 before sequencing. Sequencing was performed with triplicate samples on the Novaseq targeting 40M reads. The read counts were normalized with respect to library size and transformed using variance stabilizing transformation (VST). The resulted data was used as input to Gene Set Enrichment Analysis (GSEA) with respect to Hallmark gene sets database. Log2 Ratio of Classes was selected as the gene ranking metric, and gene set was used as permutation type. Other parameters were kept as the default values. The enrichment plots of interest were displayed.
- **Reverse Phase Protein Array (RPPA)**
 - Triplicate samples of each resistant/parental cell line were prepared for RPPA analysis. Sample analysis across a total of 485 antibodies was carried out at the MD Anderson RPPA core facility.
 - Bidirectionally Median Centering normalized, and batch corrected RPPA data in log2 scale was provided by RPPA core at MD Anderson Cancer Center. Differential protein expression analysis between resistant and parental lines was performed using *lmFit* and *eBays* methods from *limma* Bioconductor package. Common proteins of interest with adjusted p-value ≤ 0.05 in all resistant lines together with N-Ras, which is significant in the 4A8 cell line only, were row mean centered and displayed in the heatmap. The hierarchical clusters were constructed with Pearson correlation dissimilarity measure and complete agglomeration method using *ComplexHeatmap* Bioconductor package.

Results

Figure 1: Generation and characterization of cell lines with acquired ARV-471 resistance

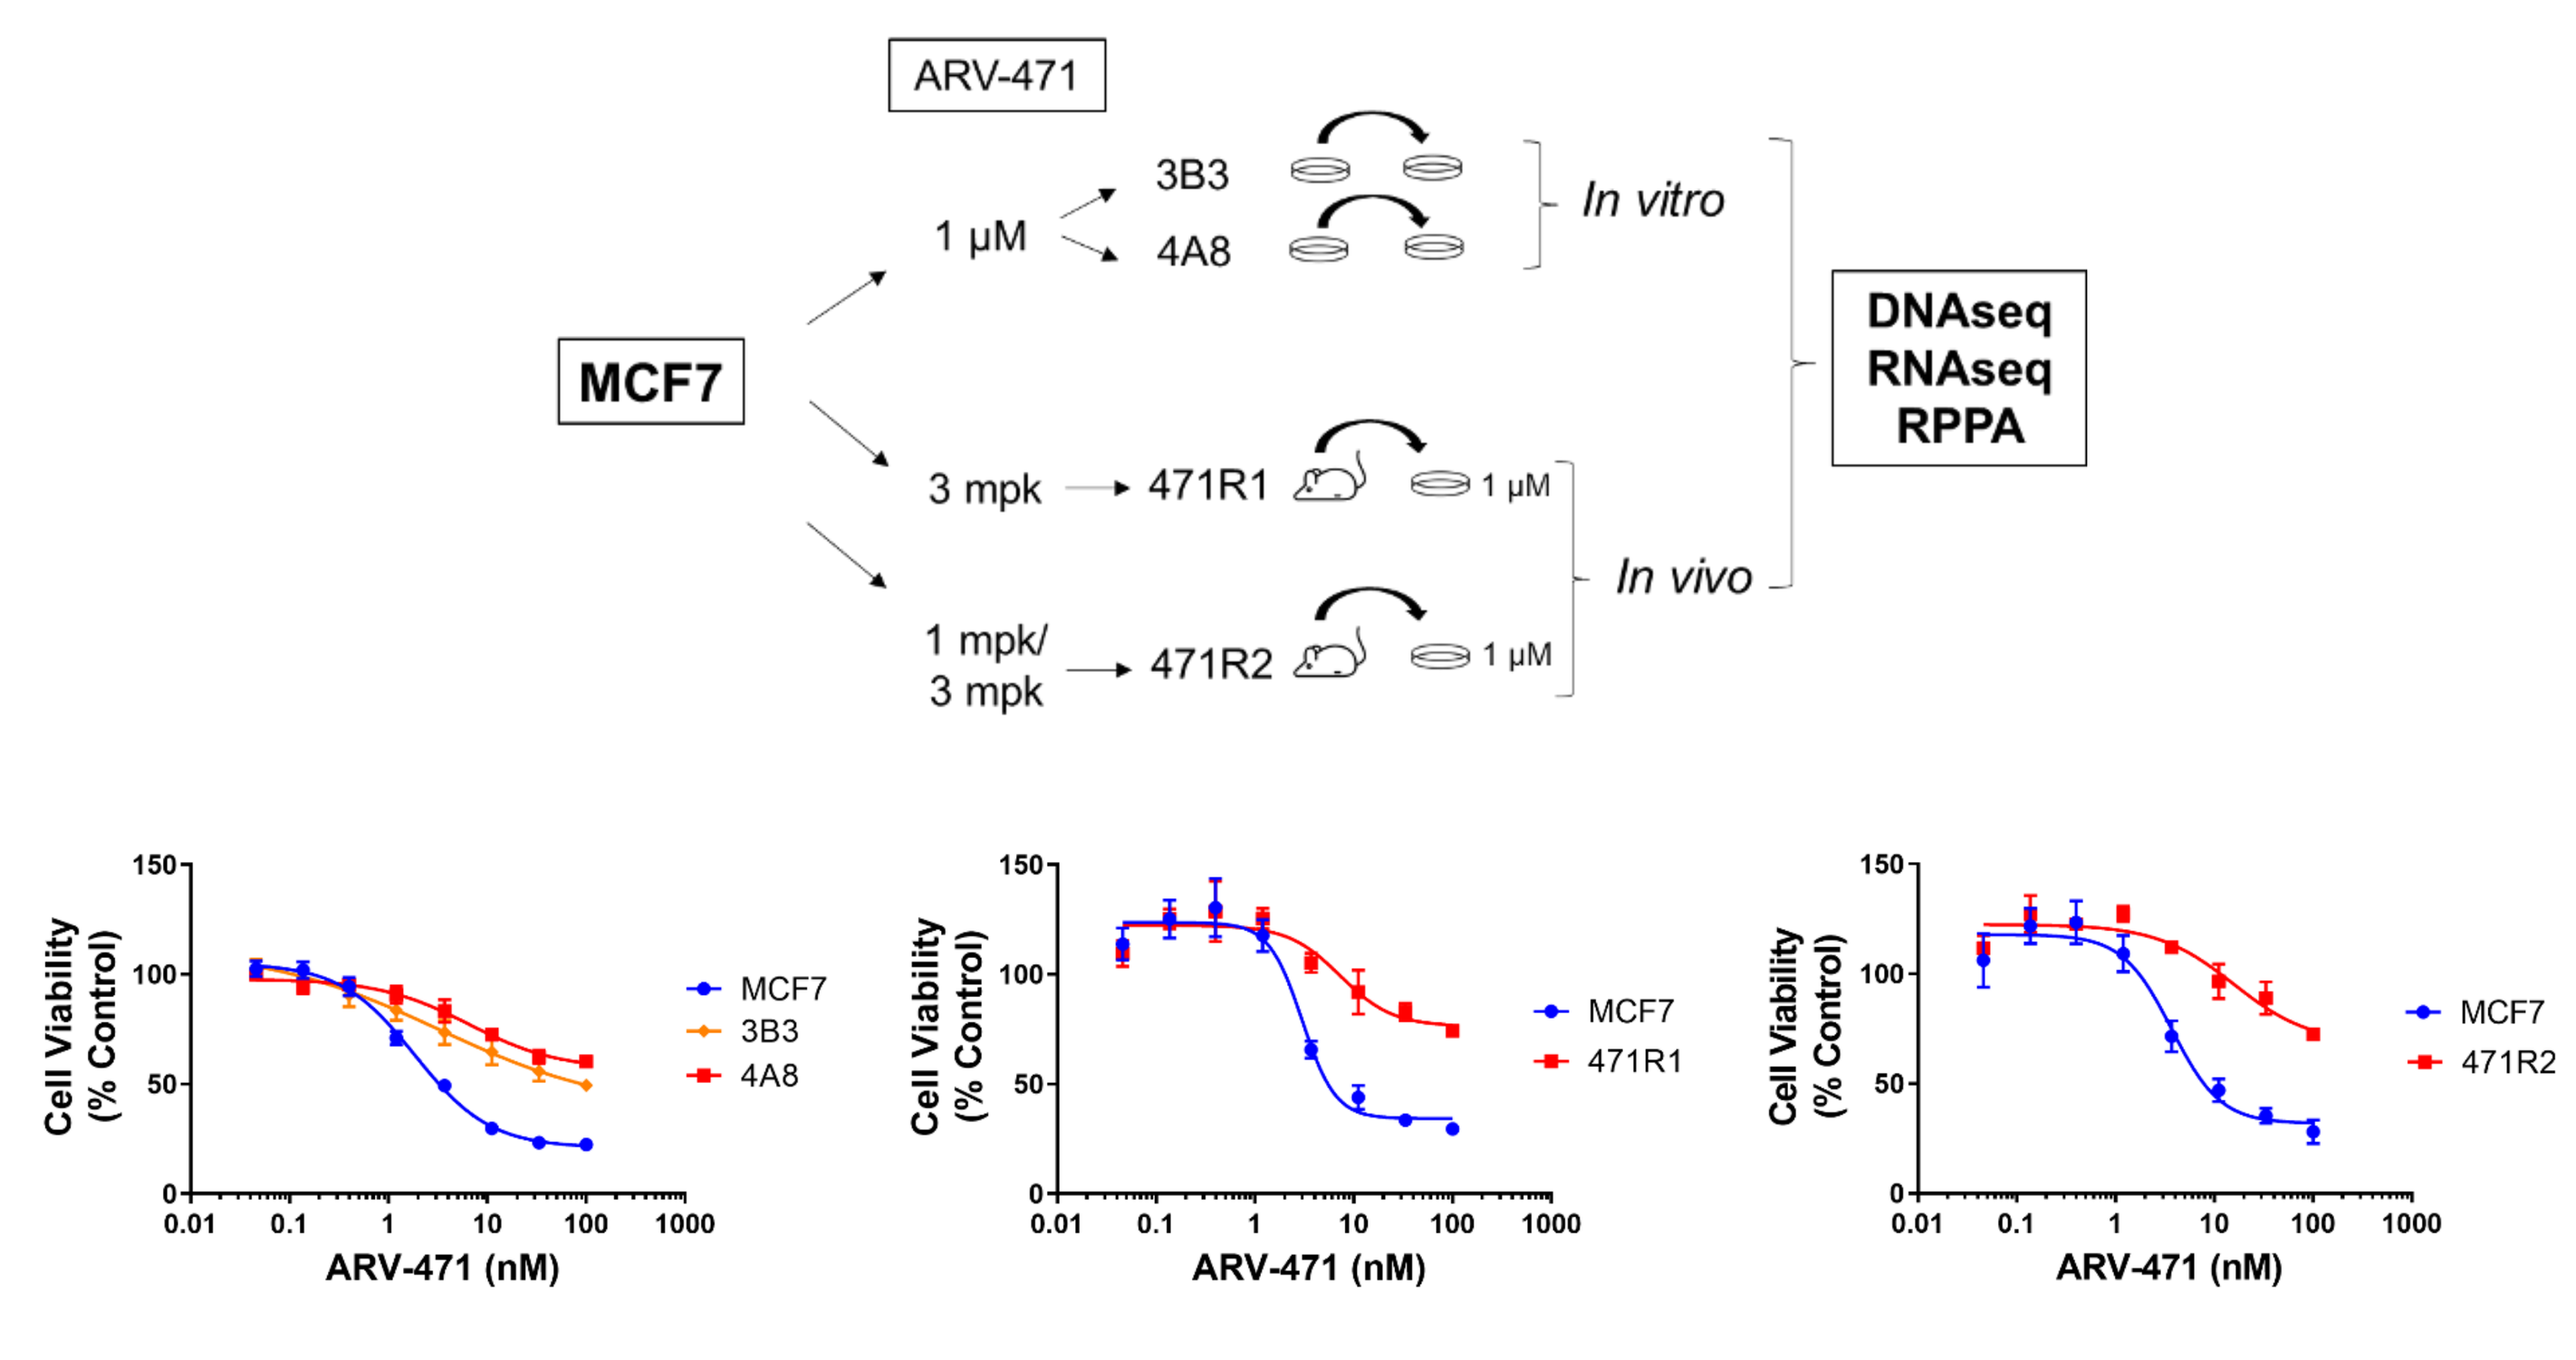
ARV-471
MCF7
1 µM
3B3
4A8
In vitro
3 mpk
471R1
1 mpk/
3 mpk
471R2
1 µM
In vivo
DNAseq
RNAseq
RPPA
Cell Viability (% Control)
ARV-471 (nM)
MCF7
3B3
4A8
471R1
471R2

mpk=milligrams per kilogram; nM=nanomolar; µM=micromolar; RPPA=Reverse Phase Protein Array.

Figure 2: Downregulation of ER protein and ER response gene expression in ARV-471 resistant cells

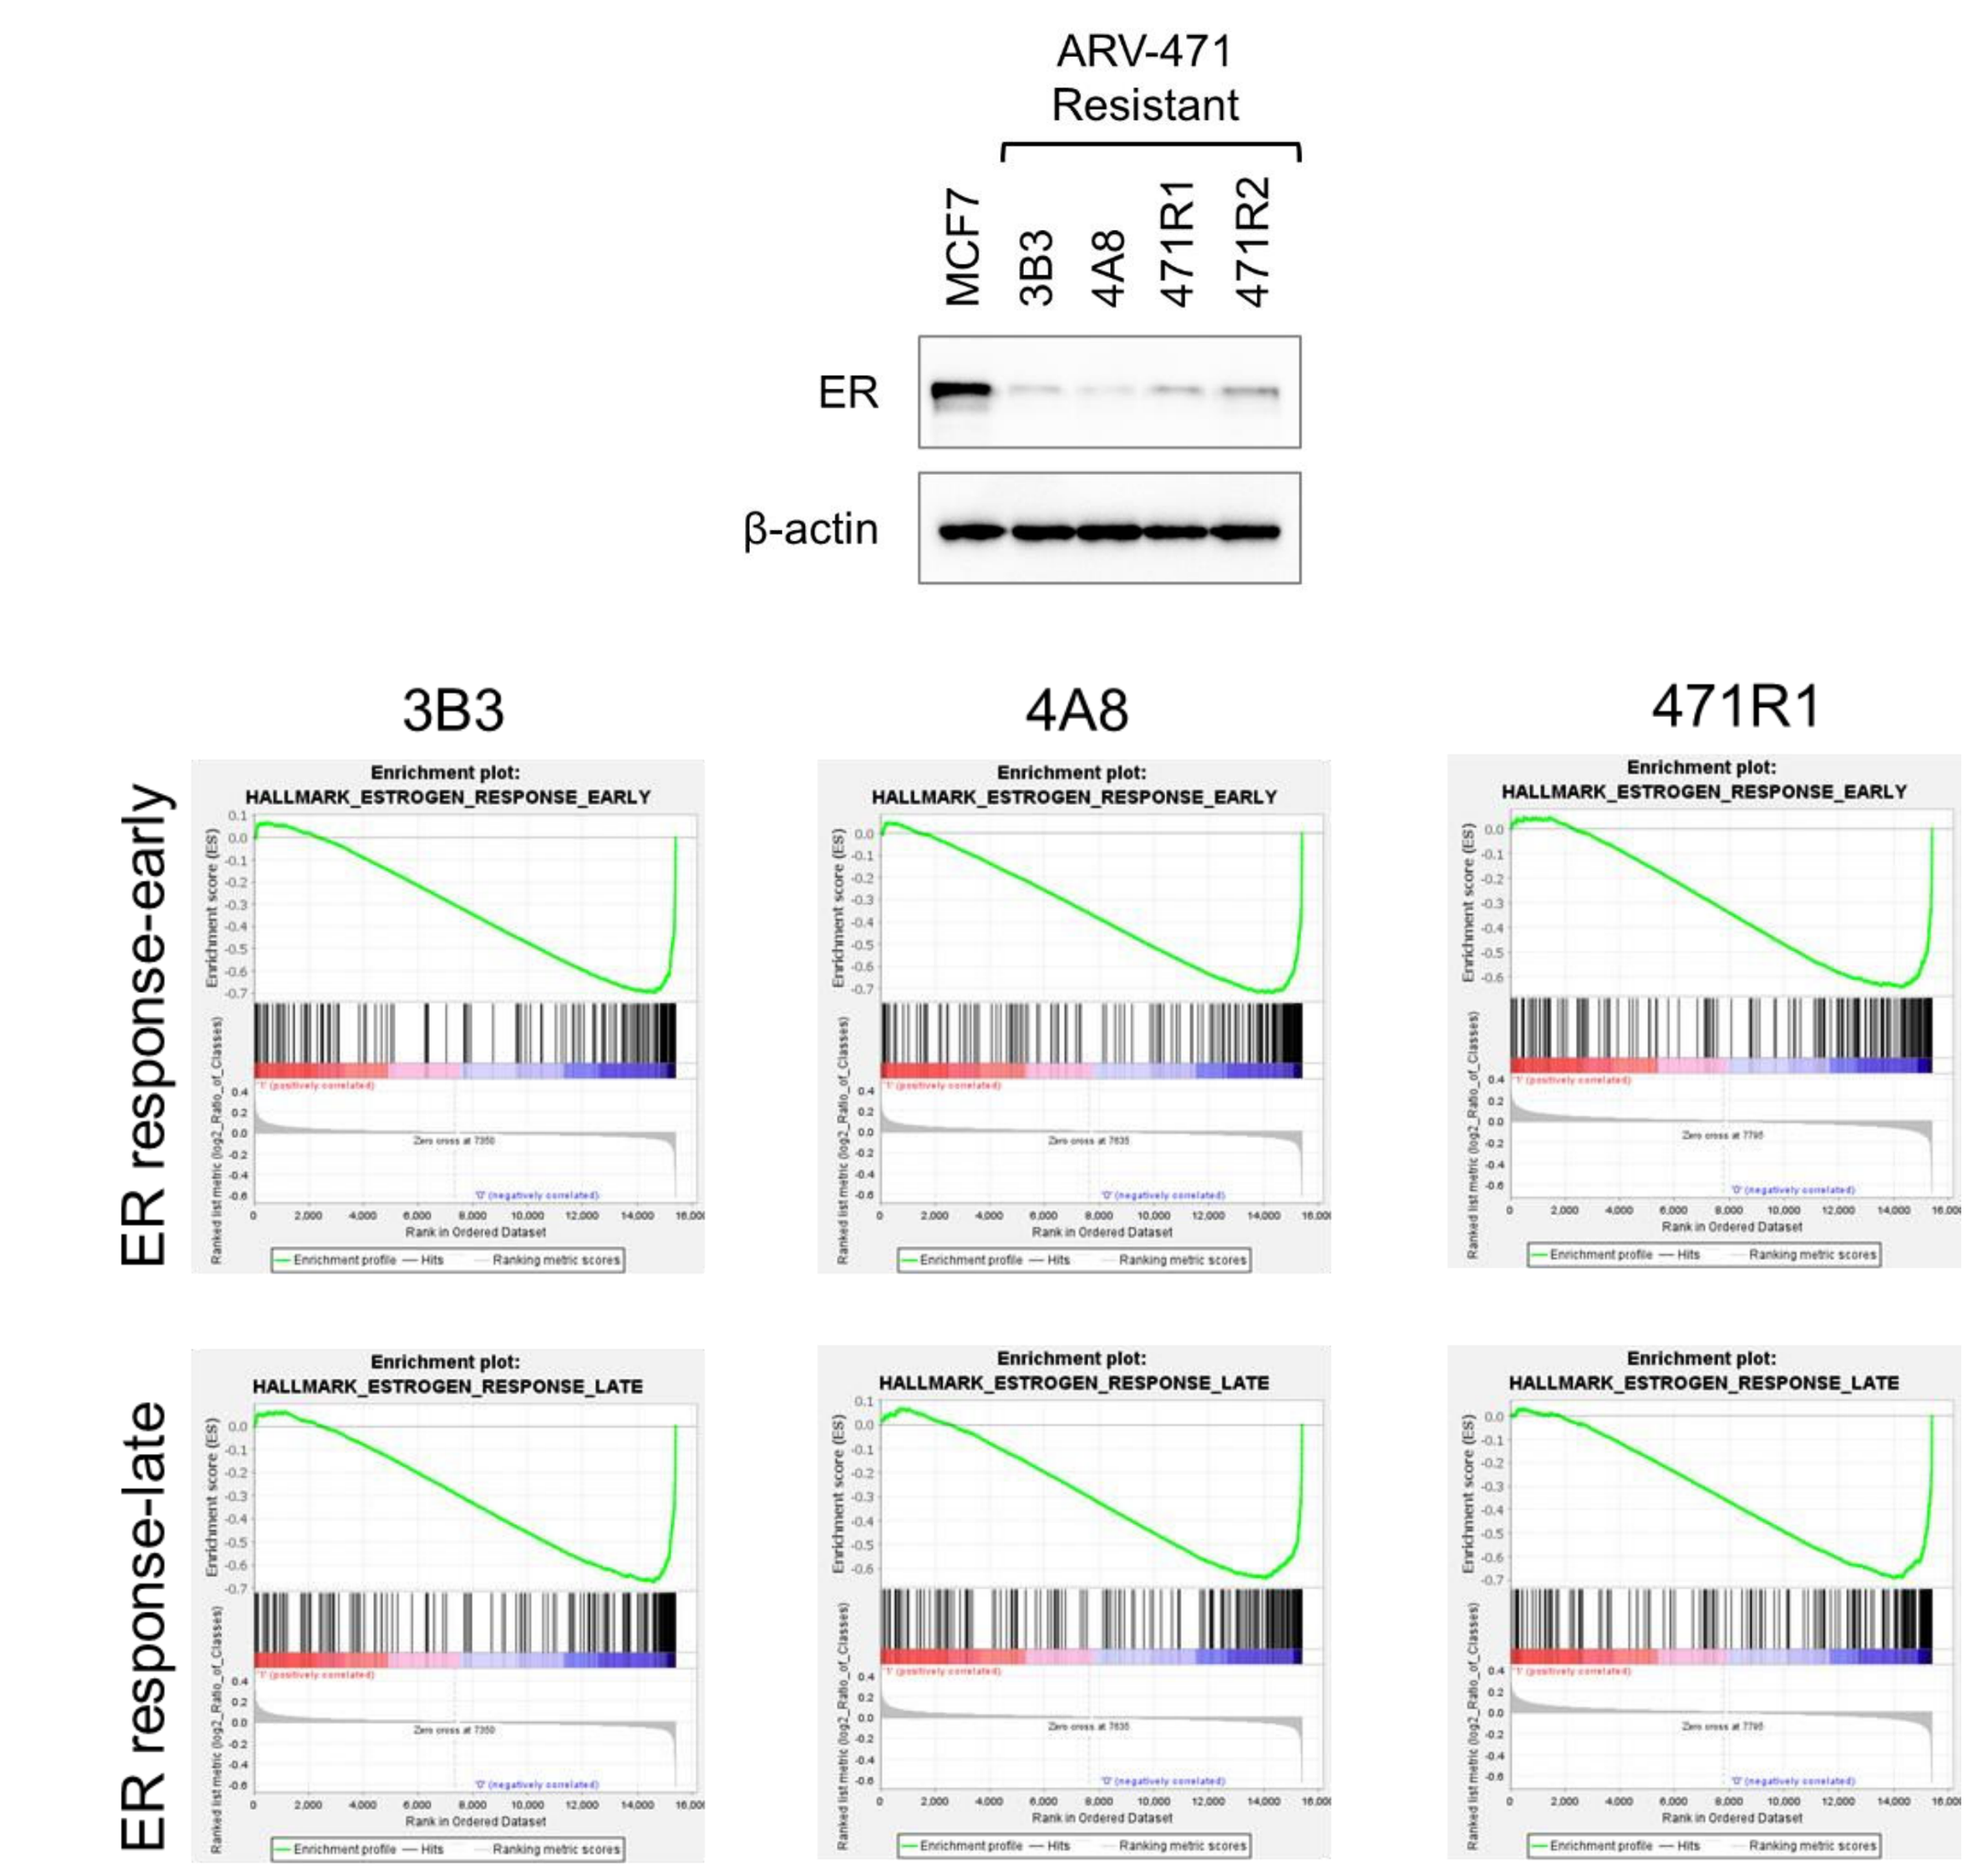
ARV-471
Resistant
MCF7
3B3
4A8
471R1
471R2
ER
β-actin
3B3
4A8
471R1
ER response-early
ER response-late
HALLMARK_ESTROGEN_RESPONSE_EARLY
HALLMARK_ESTROGEN_RESPONSE_LATE

Table 1. Whole exome sequencing of ARV-471 resistant cell lines

Cell line	Genomic alterations
3B3	none
4A8	*NRAS* gain
471R1	none

- ***ESR1* and *CRBN* alterations were not identified**
- ***NRAS* copy number gain was identified in one cell line (4A8)**

ESR1: Estrogen Receptor 1; CRBN: cereblon; NRAS: Neuroblastoma RAS viral oncogene homolog

Figure 3: Functional proteomics identified HER family, NRAS and MAPK/AKT signaling upregulation in ARV-471 resistant cells

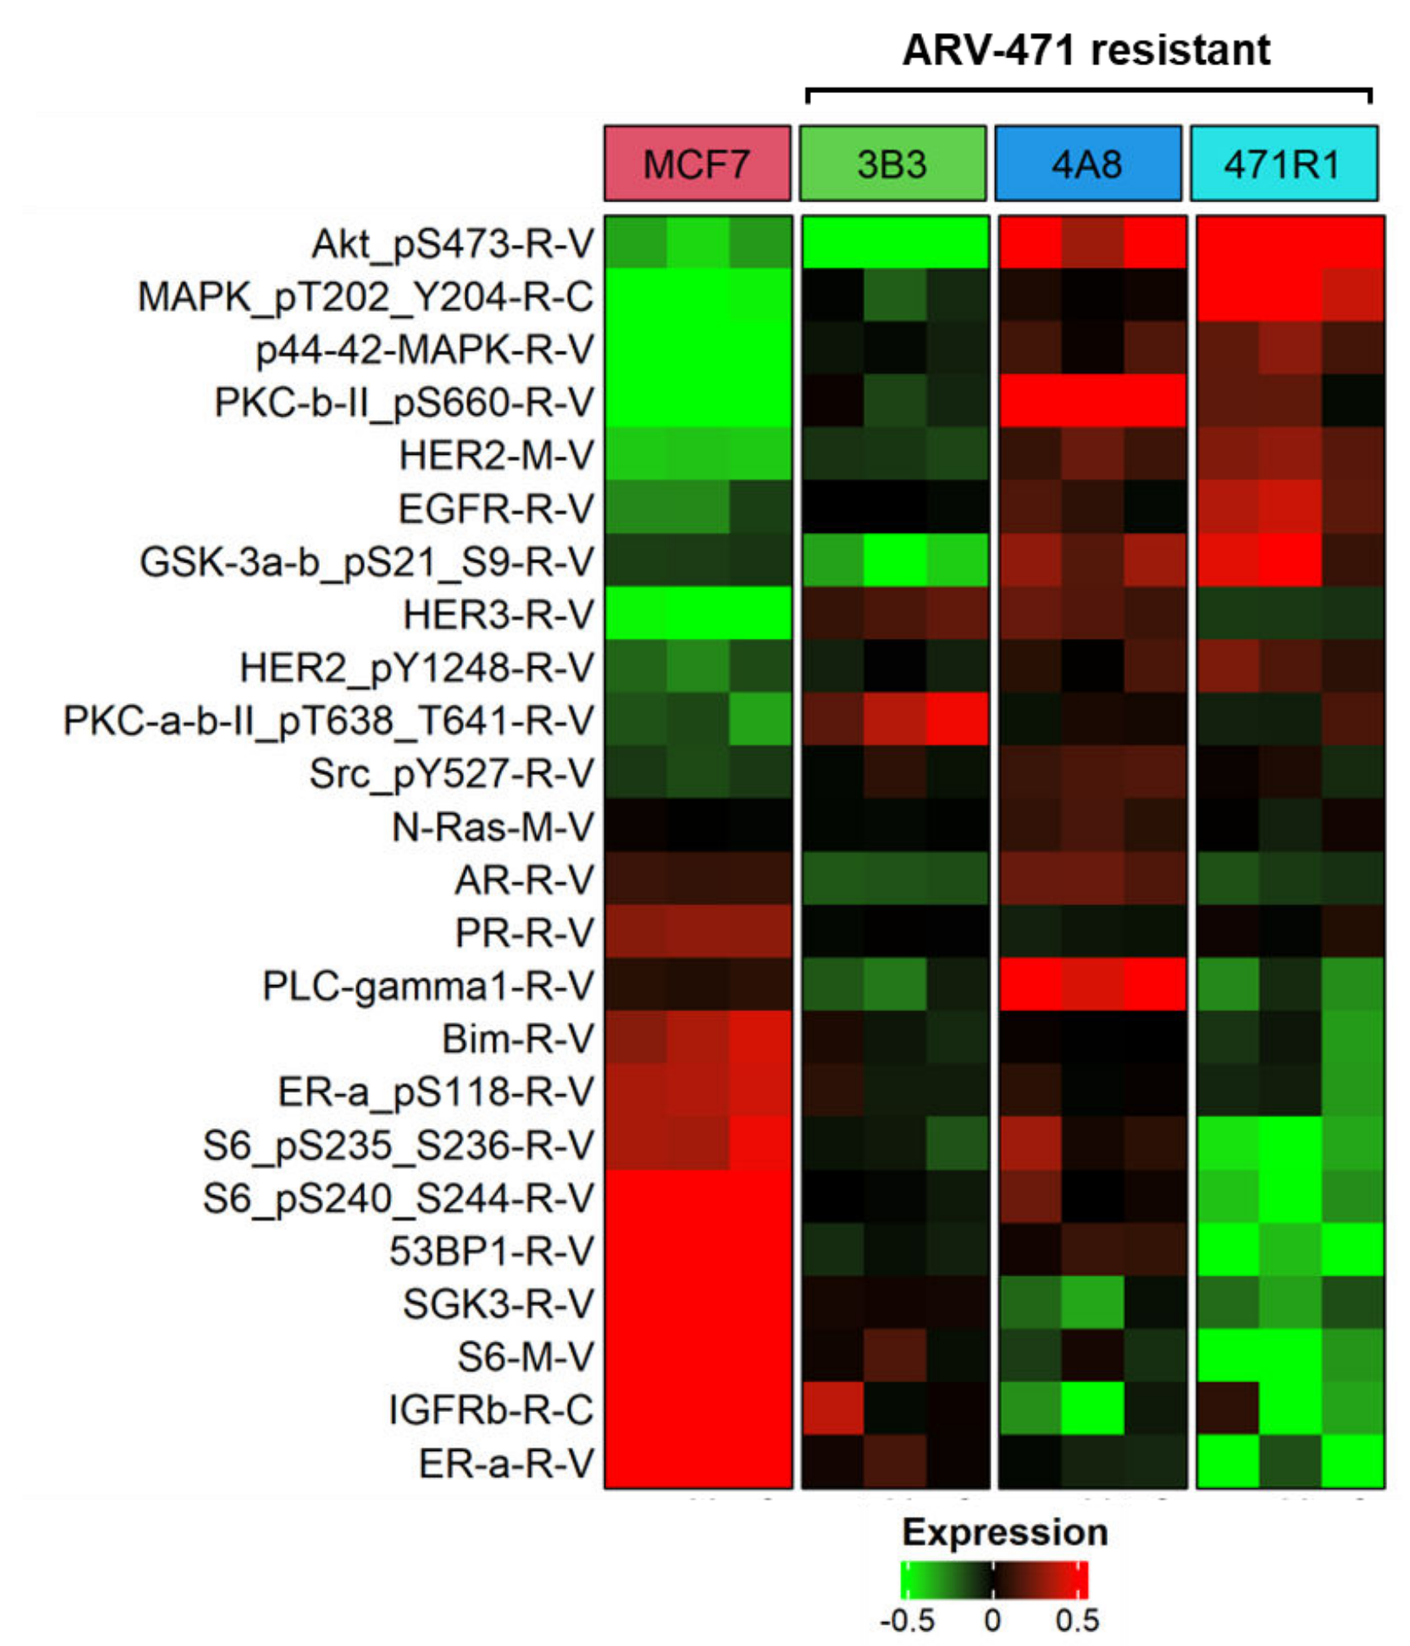
ARV-471 resistant
MCF7
3B3
4A8
471R1
Akt_pS473-R-V
MAPK_pT202_Y204-R-C
p44-42-MAPK-R-V
PKC-b-II_pS660-R-V
HER2-M-V
EGFR-R-V
GSK-3a-b_pS21_S9-R-V
HER3-R-V
HER2_pY1248-R-V
PKC-a-b-II_pT638_T641-R-V
Src_pY527-R-V
N-Ras-M-V
AR-R-V
PR-R-V
PLC-gamma1-R-V
Bim-R-V
ER-a_pS118-R-V
S6_pS235_S236-R-V
S6_pS240_S244-R-V
53BP1-R-V
SGK3-R-V
S6-M-V
IGFRb-R-C
ER-a-R-V
Expression
-0.5
0
0.5

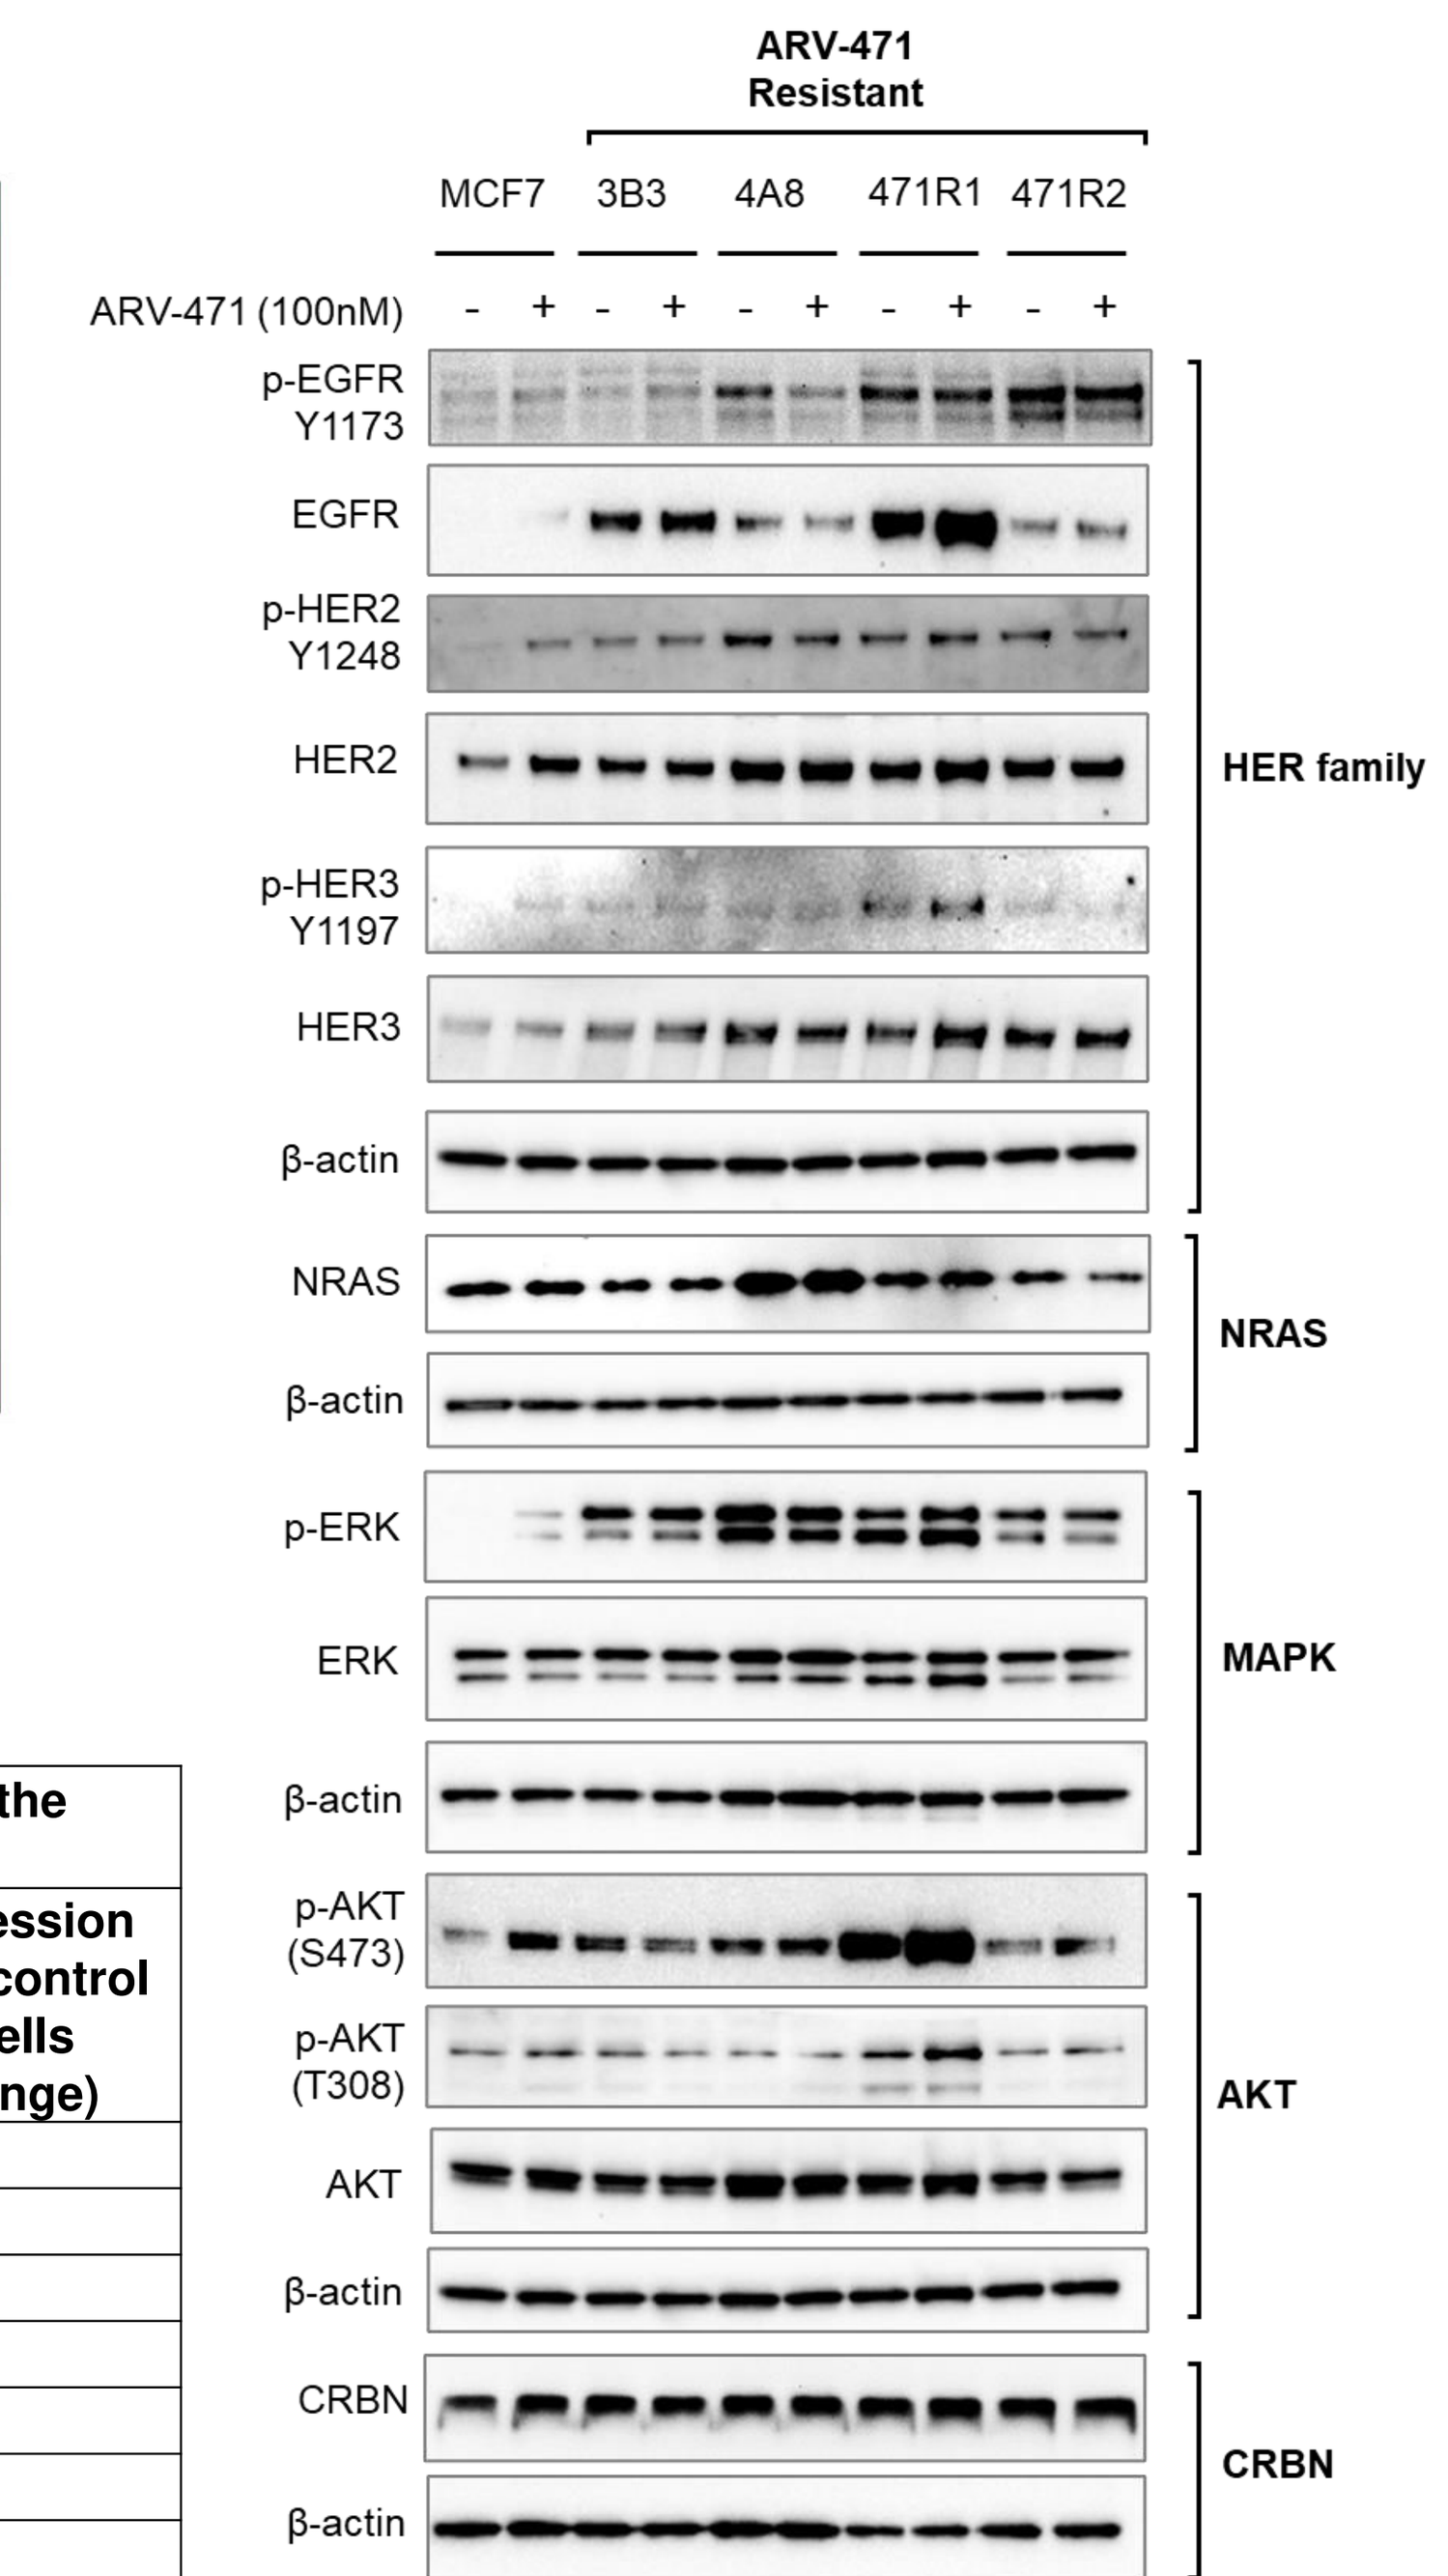
ARV-471
Resistant
MCF7
3B3
4A8
471R1
471R2
ARV-471 (100nM)
p-EGFR Y1173
EGFR
p-HER2 Y1248
HER2
p-HER3 Y1197
HER3
β-actin
NRAS
p-ERK
ERK
p-AKT (S473)
p-AKT (T308)
AKT
CRBN
HER family
NRAS
MAPK
AKT
CRBN

	Upregulation of HER family ligands at the RNA level		
	Ligand	**ARV-471-resistant cell line**	**RNA expression relative to control MCF7 cells (fold change)**
EGFR	TGFA	3B3	4.50
EGFR	TGFA	4A8	1.48
EGFR	TGFA	471R1	1.52
EGFR / HER4	HB-EGF	3B3	1.62
EGFR / HER4	HB-EGF	4A8	0.68
EGFR / HER4	HB-EGF	471R1	9.19
HER4	NRG3	3B3	2.33
HER4	NRG3	4A8	12.13
HER4	NRG3	471R1	2.36

nM: nanomolar, CRBN: cereblon

Figure 4: Overexpression of EGFR and NRAS confers resistance to ARV-471 in MCF7 cells

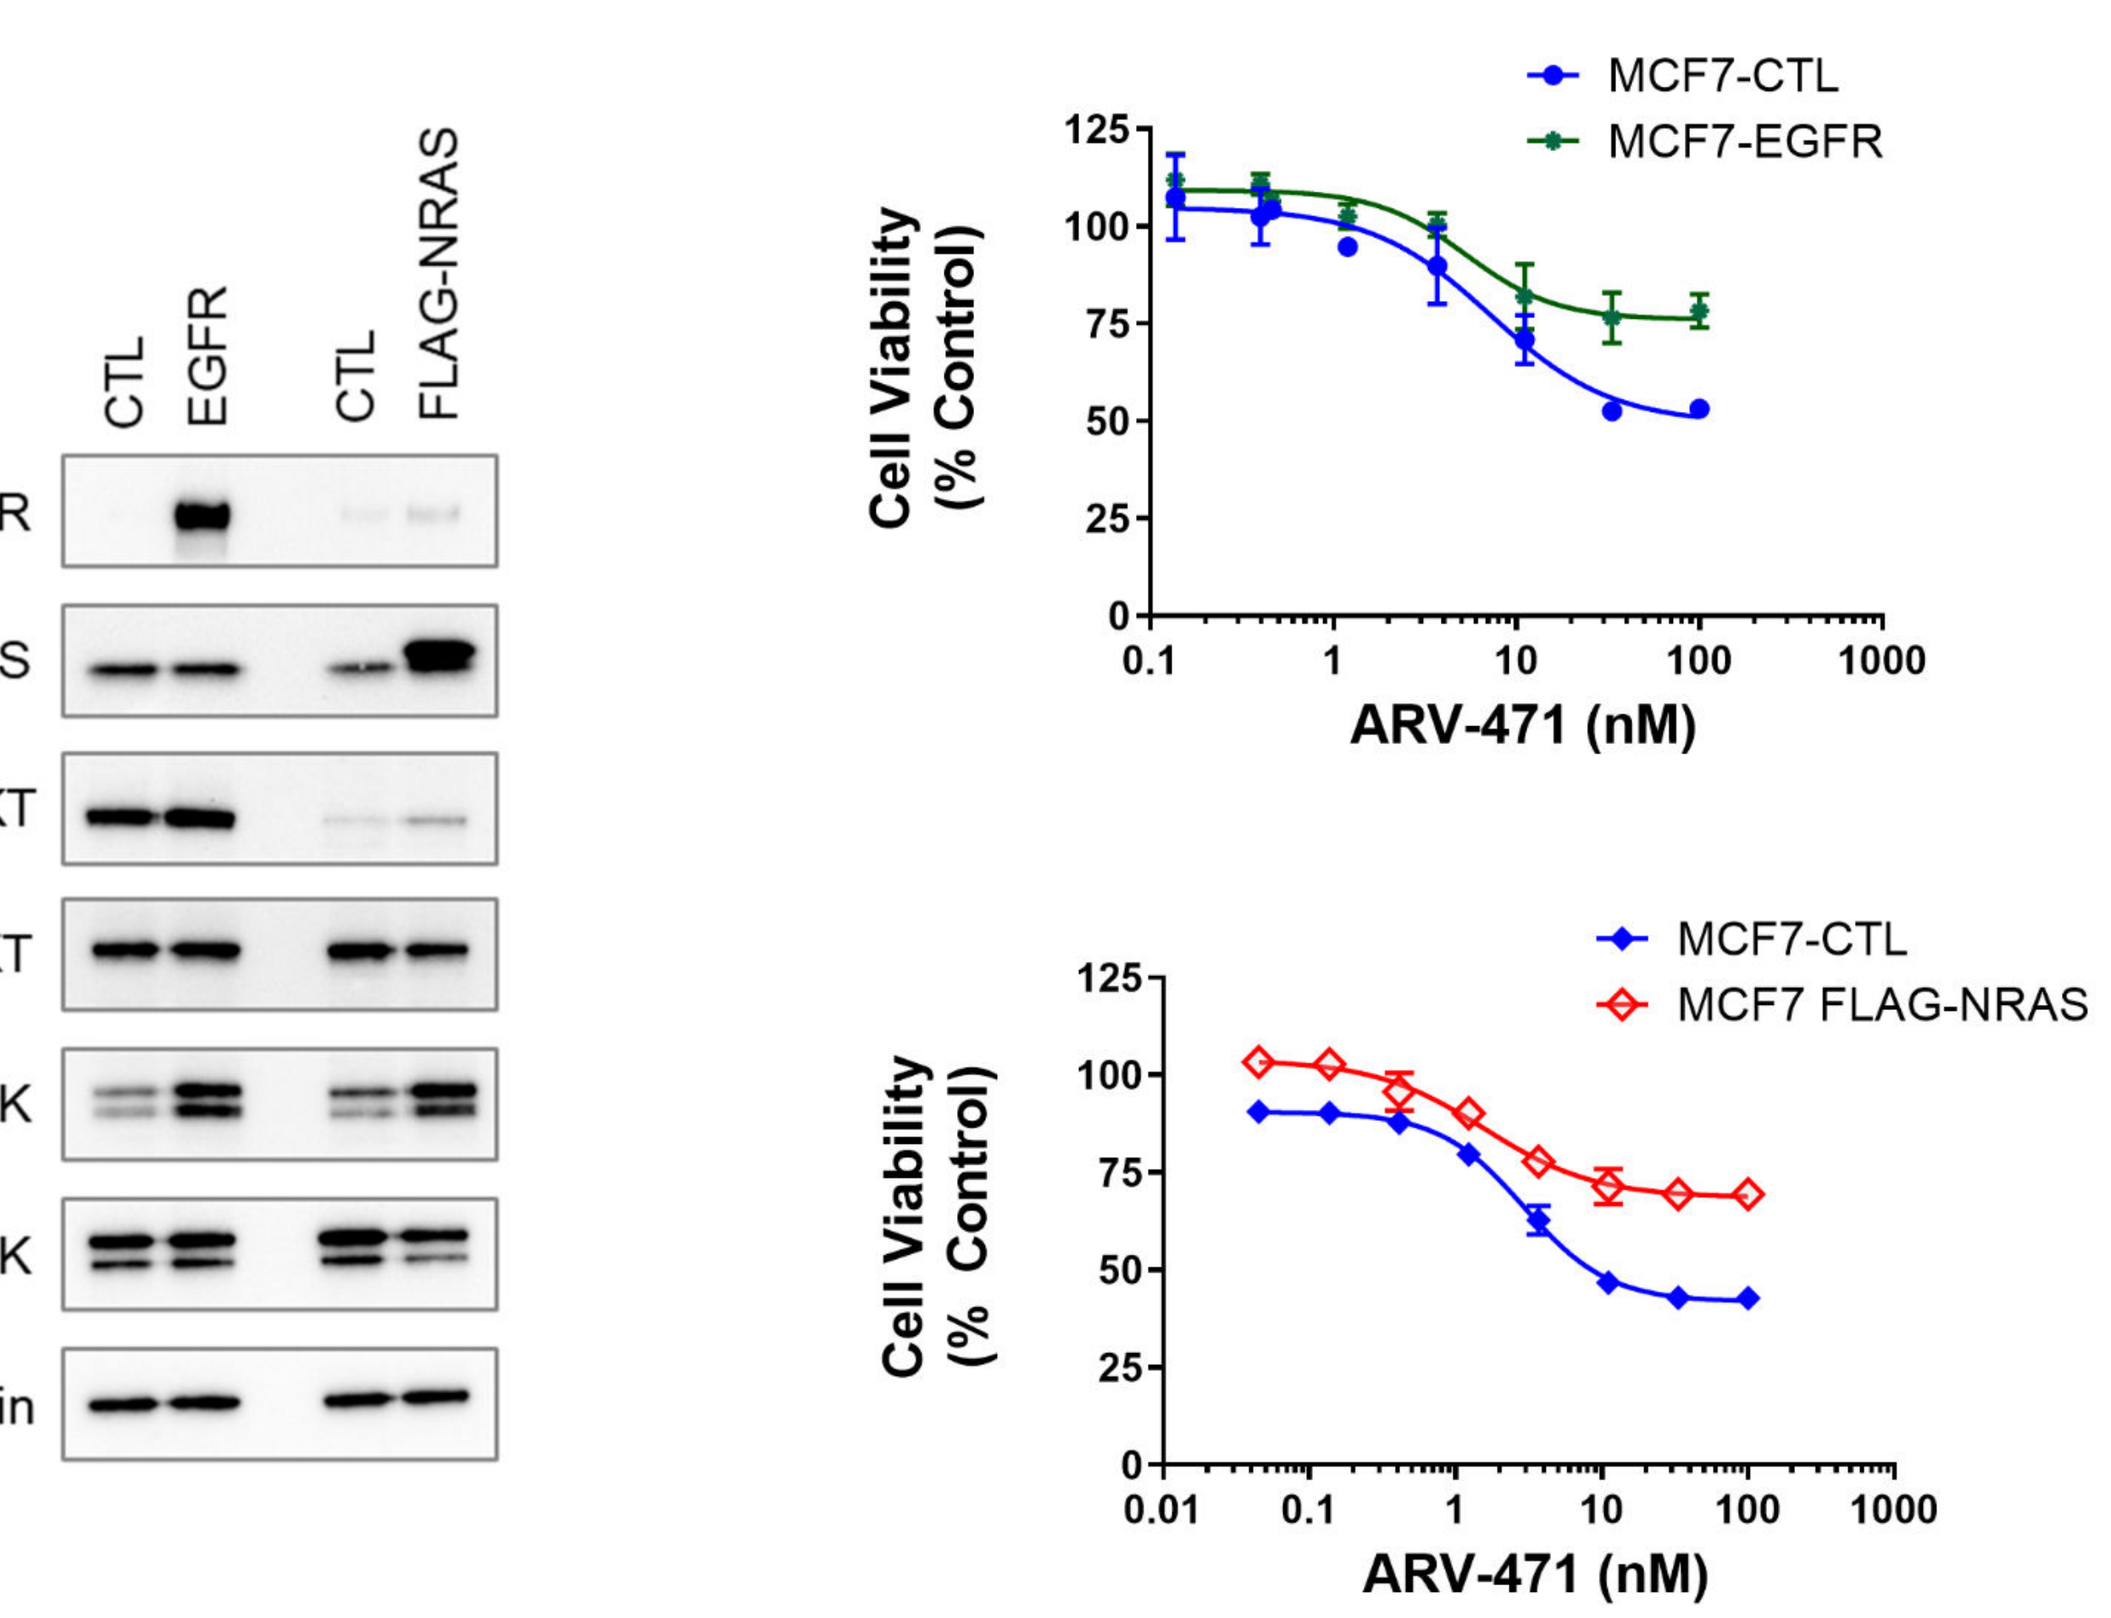
CTL
EGFR
CTL
FLAG-NRAS
EGFR
NRAS
p-AKT
AKT
p-ERK
ERK
β-Actin
MCF7-CTL
MCF7-EGFR
MCF7 FLAG-NRAS
Cell Viability (% Control)
ARV-471 (nM)

MCF7 cells were transduced with either a control empty lentivirus (CTL) or lentiviruses expressing human EGFR or NRAS.
EGFR: Epidermal Growth Factor Receptor, NRAS: neuroblastoma ras viral oncogene homolog, CTL: Control, nM: nanomolar

Figure 5: ARV-471 resistant cells with EGFR pathway upregulation are sensitive to EGFR, pan-HER and MAPK pathway inhibitors

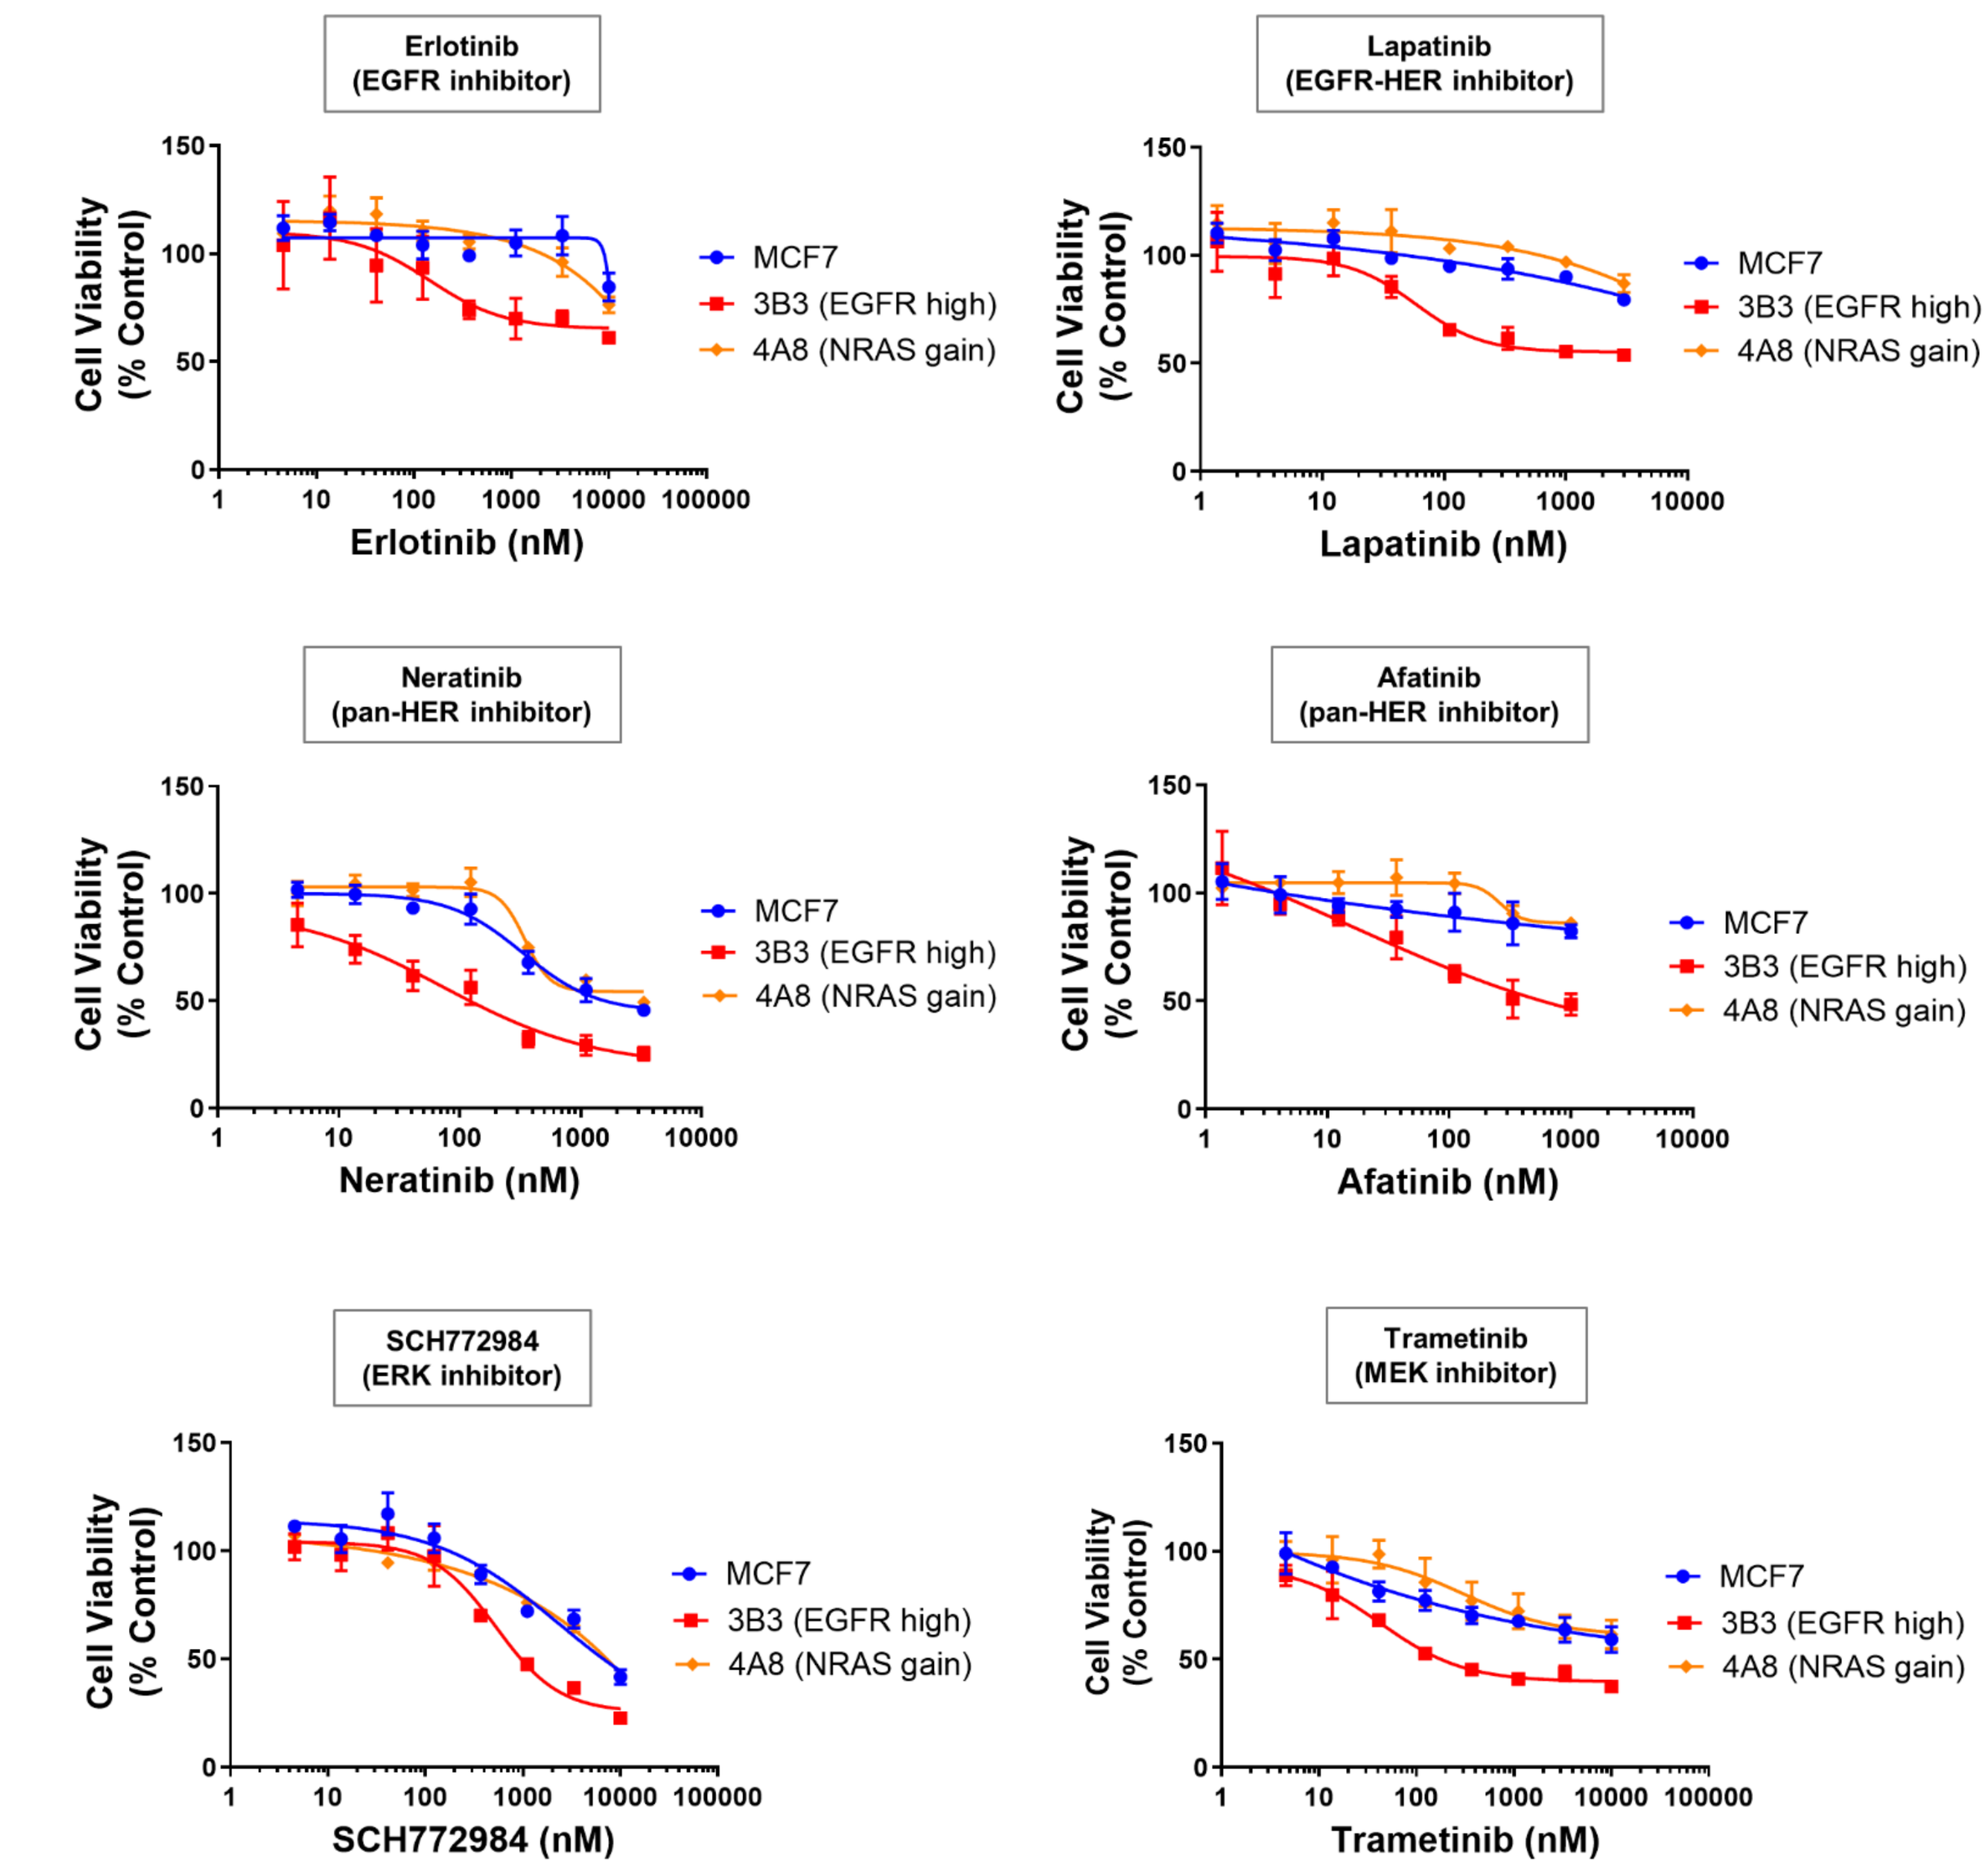
Erlotinib
(EGFR inhibitor)
Lapatinib
(EGFR-HER inhibitor)
Neratinib
(pan-HER inhibitor)
Afatinib
(pan-HER inhibitor)
SCH772984
(ERK inhibitor)
Trametinib
(MEK inhibitor)
Cell Viability (% Control)
MCF7
3B3 (EGFR high)
4A8 (NRAS gain)
Erlotinib (nM)
Lapatinib (nM)
Neratinib (nM)
Afatinib (nM)
SCH772984 (nM)
Trametinib (nM)

EGFR: Epidermal Growth Factor Receptor, HER: Human Epidermal Growth Factor Receptor, ERK: Extracellular Signal-Regulated Kinase, MEK: Mitogen-activated protein kinase kinase, nM: nanomolar

Figure 6: ARV-471 inhibits ER signaling in the absence of degradation

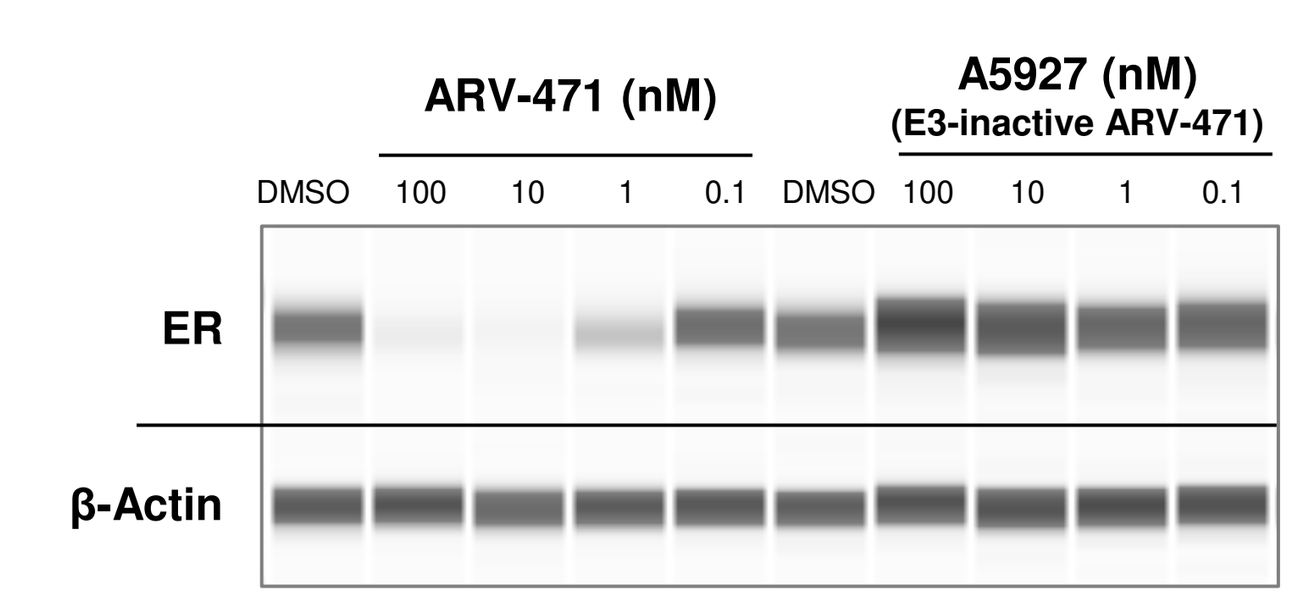
ARV-471 (nM)
A5927 (nM)
(E3-inactive ARV-471)
DMSO
100
10
1
0.1
ER
β-Actin

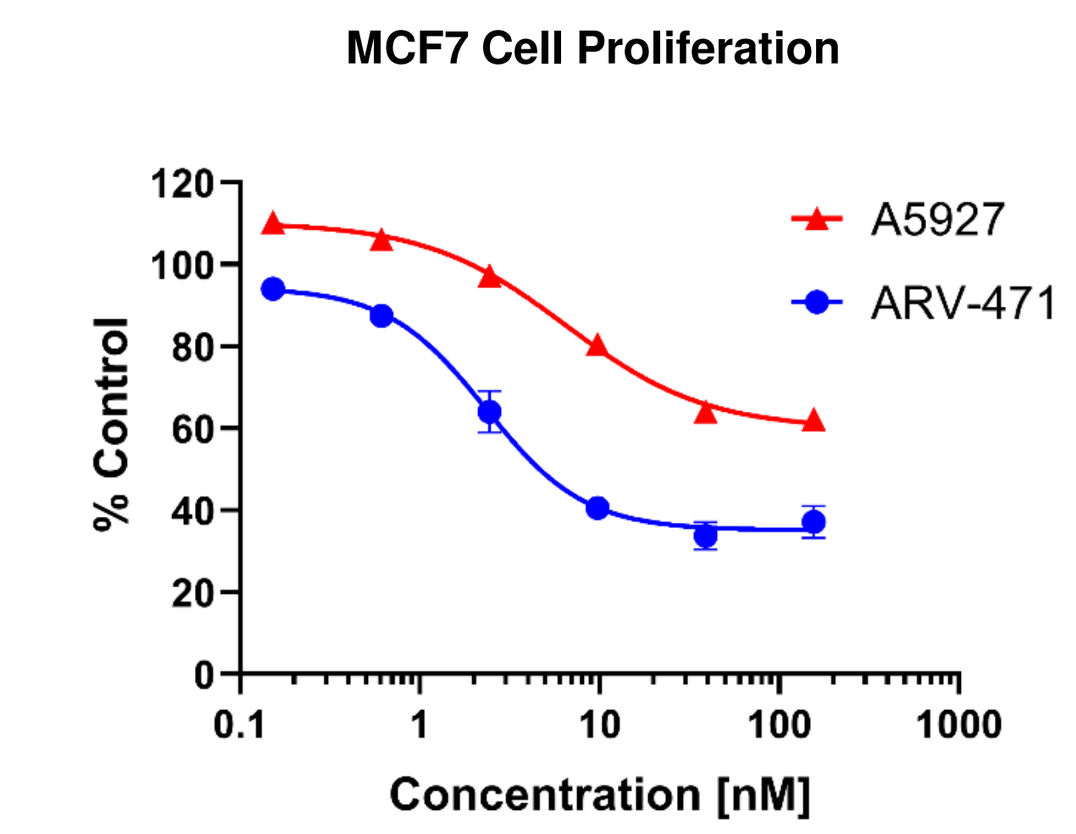
MCF7 Cell Proliferation
A5927
ARV-471
% Control
Concentration [nM]

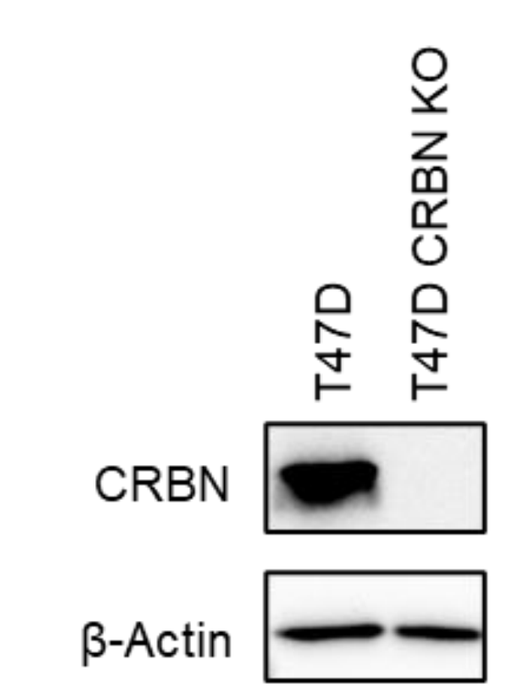
T47D
T47D CRBN KO
CRBN
β-Actin

Effect of ARV-471 on Parental and CRBN Knockout ER+ T47D Breast Cancer Cell Proliferation	
Cell Line	**ARV-471 Mean EC_{50} ± SEM (nM)**
T47D	1.7 ± 0.5
T47D CRBN KO	8.0 ± 2.5

CRBN: cereblon; KO: Knockout; SEM: Standard Error of the Mean; Mean of 3 independent experiments.